Trial in progress: a randomized, multicenter, double-blind, placebo-controlled, phase 2b study to assess the safety and efficacy of IGV-001, an autologous cell immunotherapy with antisense oligonucleotide (IMV-001) targeting IGF-1R, in newly diagnosed patients with glioblastoma

TIPS-18

Ian Y. Lee,[1] Simon Hanft,[2] Michael Schulder,[3] Kevin D. Judy,[4] Eric T. Wong,[5] J. Bradley Elder,[6] Linton T. Evans,[7] Mario Zuccarello,[8] Julian Wu,[9] Sonikpreet Aulakh,[10] Vijay Agarwal,[11] Rohan Ramakrishna,[12] Brian J. Gill,[13] Alfredo Quiñones-Hinojosa,[14] Cameron Brennan,[15] Brad E. Zacharia,[16] Carlos Eduardo Silva Correia,[17] Madhavi Diwanji,[18] Gregory K. Pennock,[18] Charles Scott,[18] Raul Perez-Olle,[18] David W. Andrews,[18] John A. Boockvar[19]

[1]Henry Ford Health System, Detroit, MI, USA; [2]Westchester Medical Center, Valhalla, NY, USA; [3]Northwell Health at North Shore University Hospital, Lake Success, NY, USA; [4]Thomas Jefferson University, Philadelphia, PA, USA; [5]Rhode Island Hospital and The Warren Alpert Medical School of Brown University, Providence, RI, USA; [6]Ohio State University, Columbus, OH, USA; [7]Dartmouth Hitchcock Medical Center, Lebanon, NH, USA; [8]University of Cincinnati Medical Center, Cincinnati, OH, USA; [9]Tufts Medical Center, Boston, MA, USA; [10]West Virginia University, Morgantown, WV, USA; [11]Montefiore Medical Center, Bronx, NY, USA; [12]Cornell University Weill Medical Center, New York, NY, USA; [13]Columbia University Medical Center, New York, NY, USA; [14]Mayo Clinic, Jacksonville, FL, USA; [15]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [16]Penn State University, Hershey, PA, USA; [17]Jersey Shore University Medical Center, Neptune City, NJ, USA;[18]Imvax, Inc., Philadelphia, PA, USA; [19]Lenox Hill Hospital, New York, NY, USA.

INTRODUCTION

- Standard-of-care (SOC) for first-line therapy in patients with newly diagnosed glioblastoma (GBM) is surgery followed by concurrent radiotherapy (RT) and temozolomide (TMZ) followed by adjuvant TMZ alone as maintenance[1]
- With SOC, overall survival (OS) was 14.6 months and progression-free survival (PFS) was 6.9 months in the Stupp trial[1]
- Insulin-like growth factor type 1 receptor (IGF-1R) is overexpressed in malignant cells, including GBM,[2] where it promotes cell growth, cell survival, and tumor progression, and is implicated in the pathophysiology of several human cancers[3-6]
- IGF-1R leads to activation of the PI3K/Akt and the Ras/Raf/MEK/MAPK signaling pathways[3,4]
- IGF-1R signaling protects cancer cells from apoptosis induced by RT and anticancer drugs[7-9]
- Downregulation of IGF-1R function provides a selective target for anticancer therapies and antitumor activity of IGF-1R inhibition has been demonstrated in preclinical studies[3,10-12]
- IGV-001 is the first product developed using Goldspire™, Imvax's proprietary platform (**Figure 1**)

Figure 1. The Goldspire™ platform

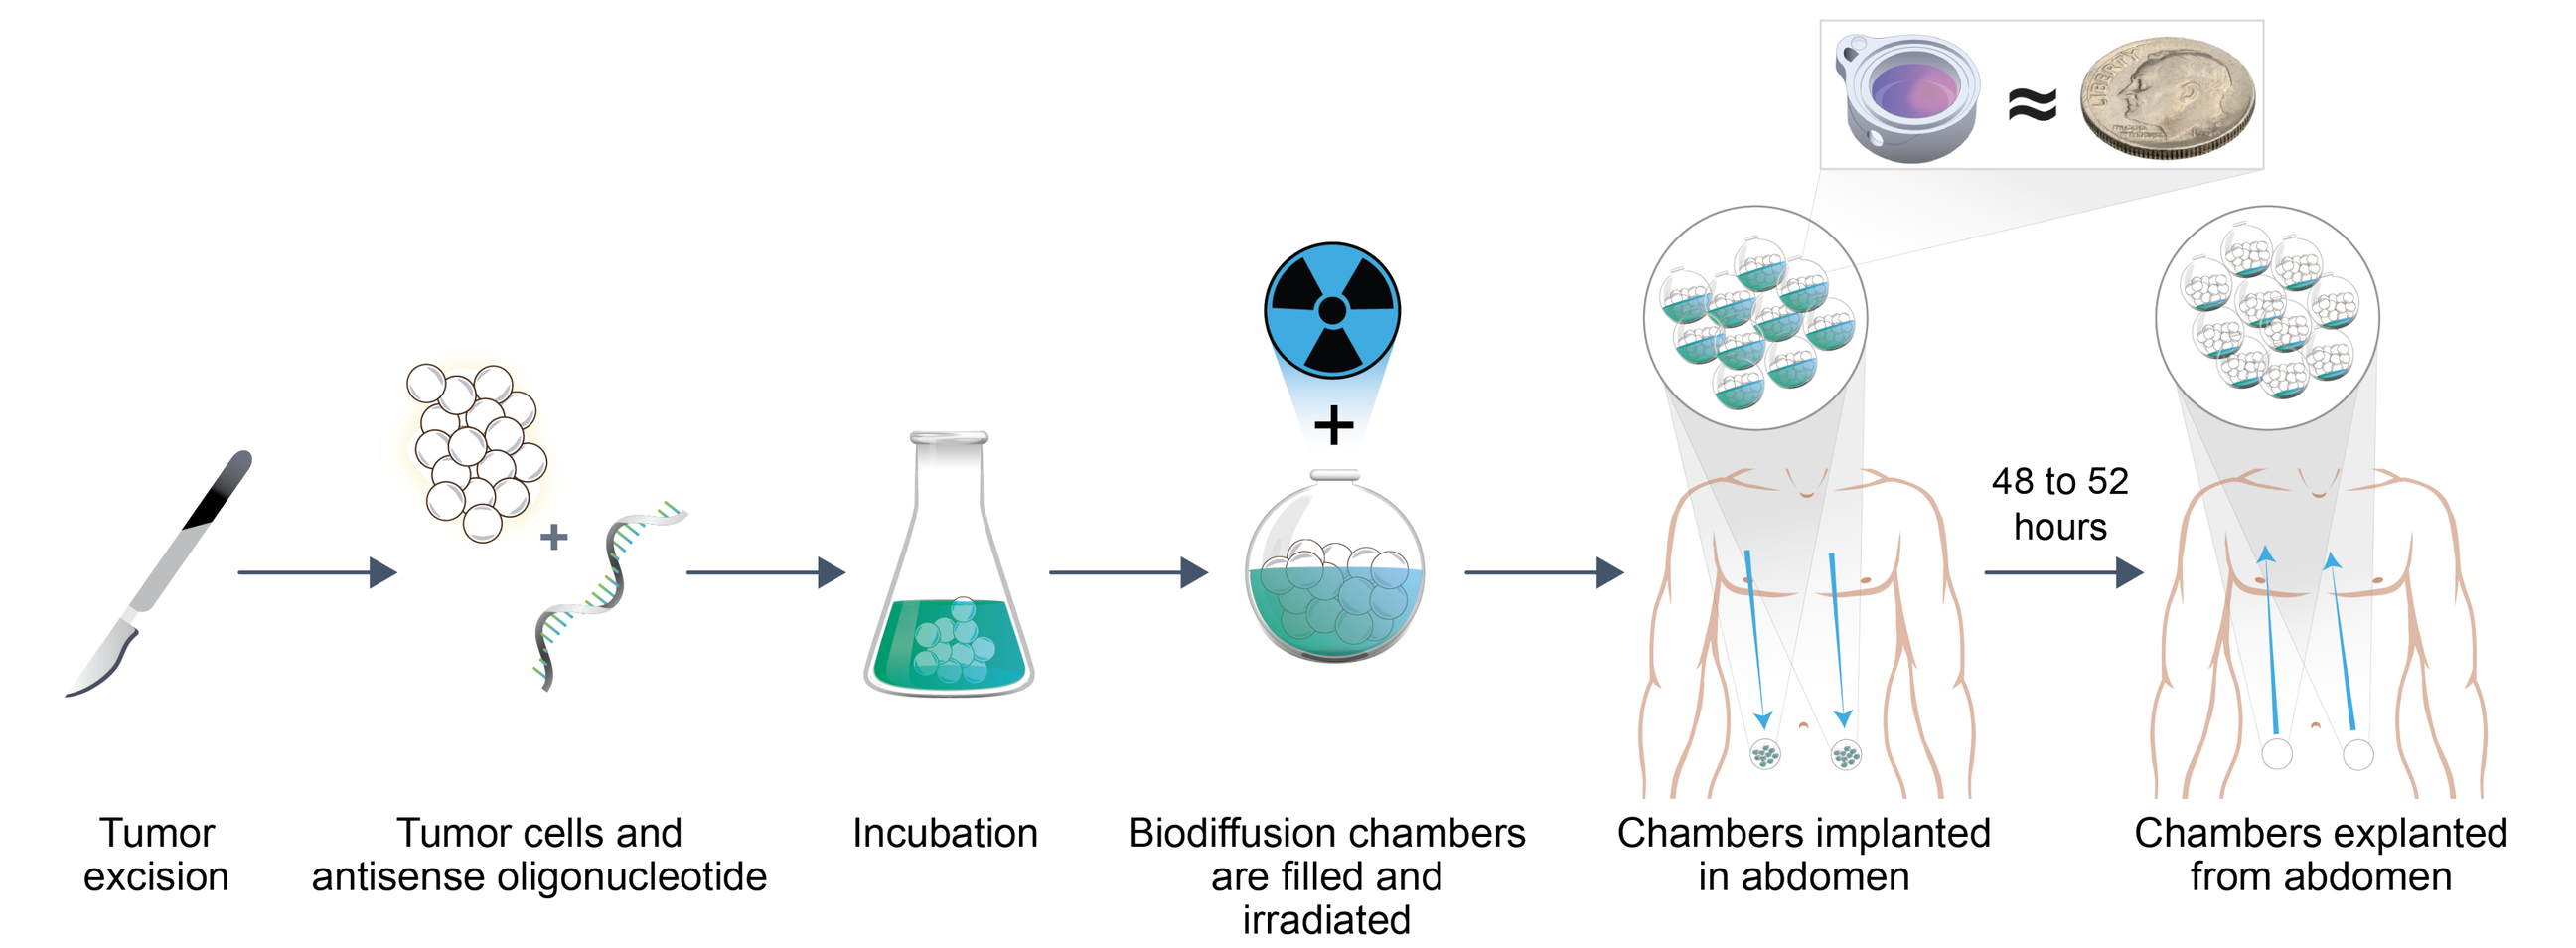

≈
+
48 to 52 hours
Tumor excision
Tumor cells and antisense oligonucleotide
Incubation
Biodiffusion chambers are filled and irradiated
Chambers implanted in abdomen
Chambers explanted from abdomen

- IGV-001 is a cellular immunotherapy combination drug product consisting of a heterogeneous mixture of autologous cells that have been isolated from resected GBM tumor tissue incubated with IMV-001, a single-stranded 18-mer antisense oligonucleotide corresponding to the 6 codons downstream from the initiating methionine codon of the IGF-1R coding sequence
 - Through its effects on IGF-1R, IMV-001 is believed to enhance antigen release and expected to activate antigen presentation (**Figure 2**)[13,14]

Figure 2. The IGV-001 manufacturing assembly and 5-stage mechanism of action

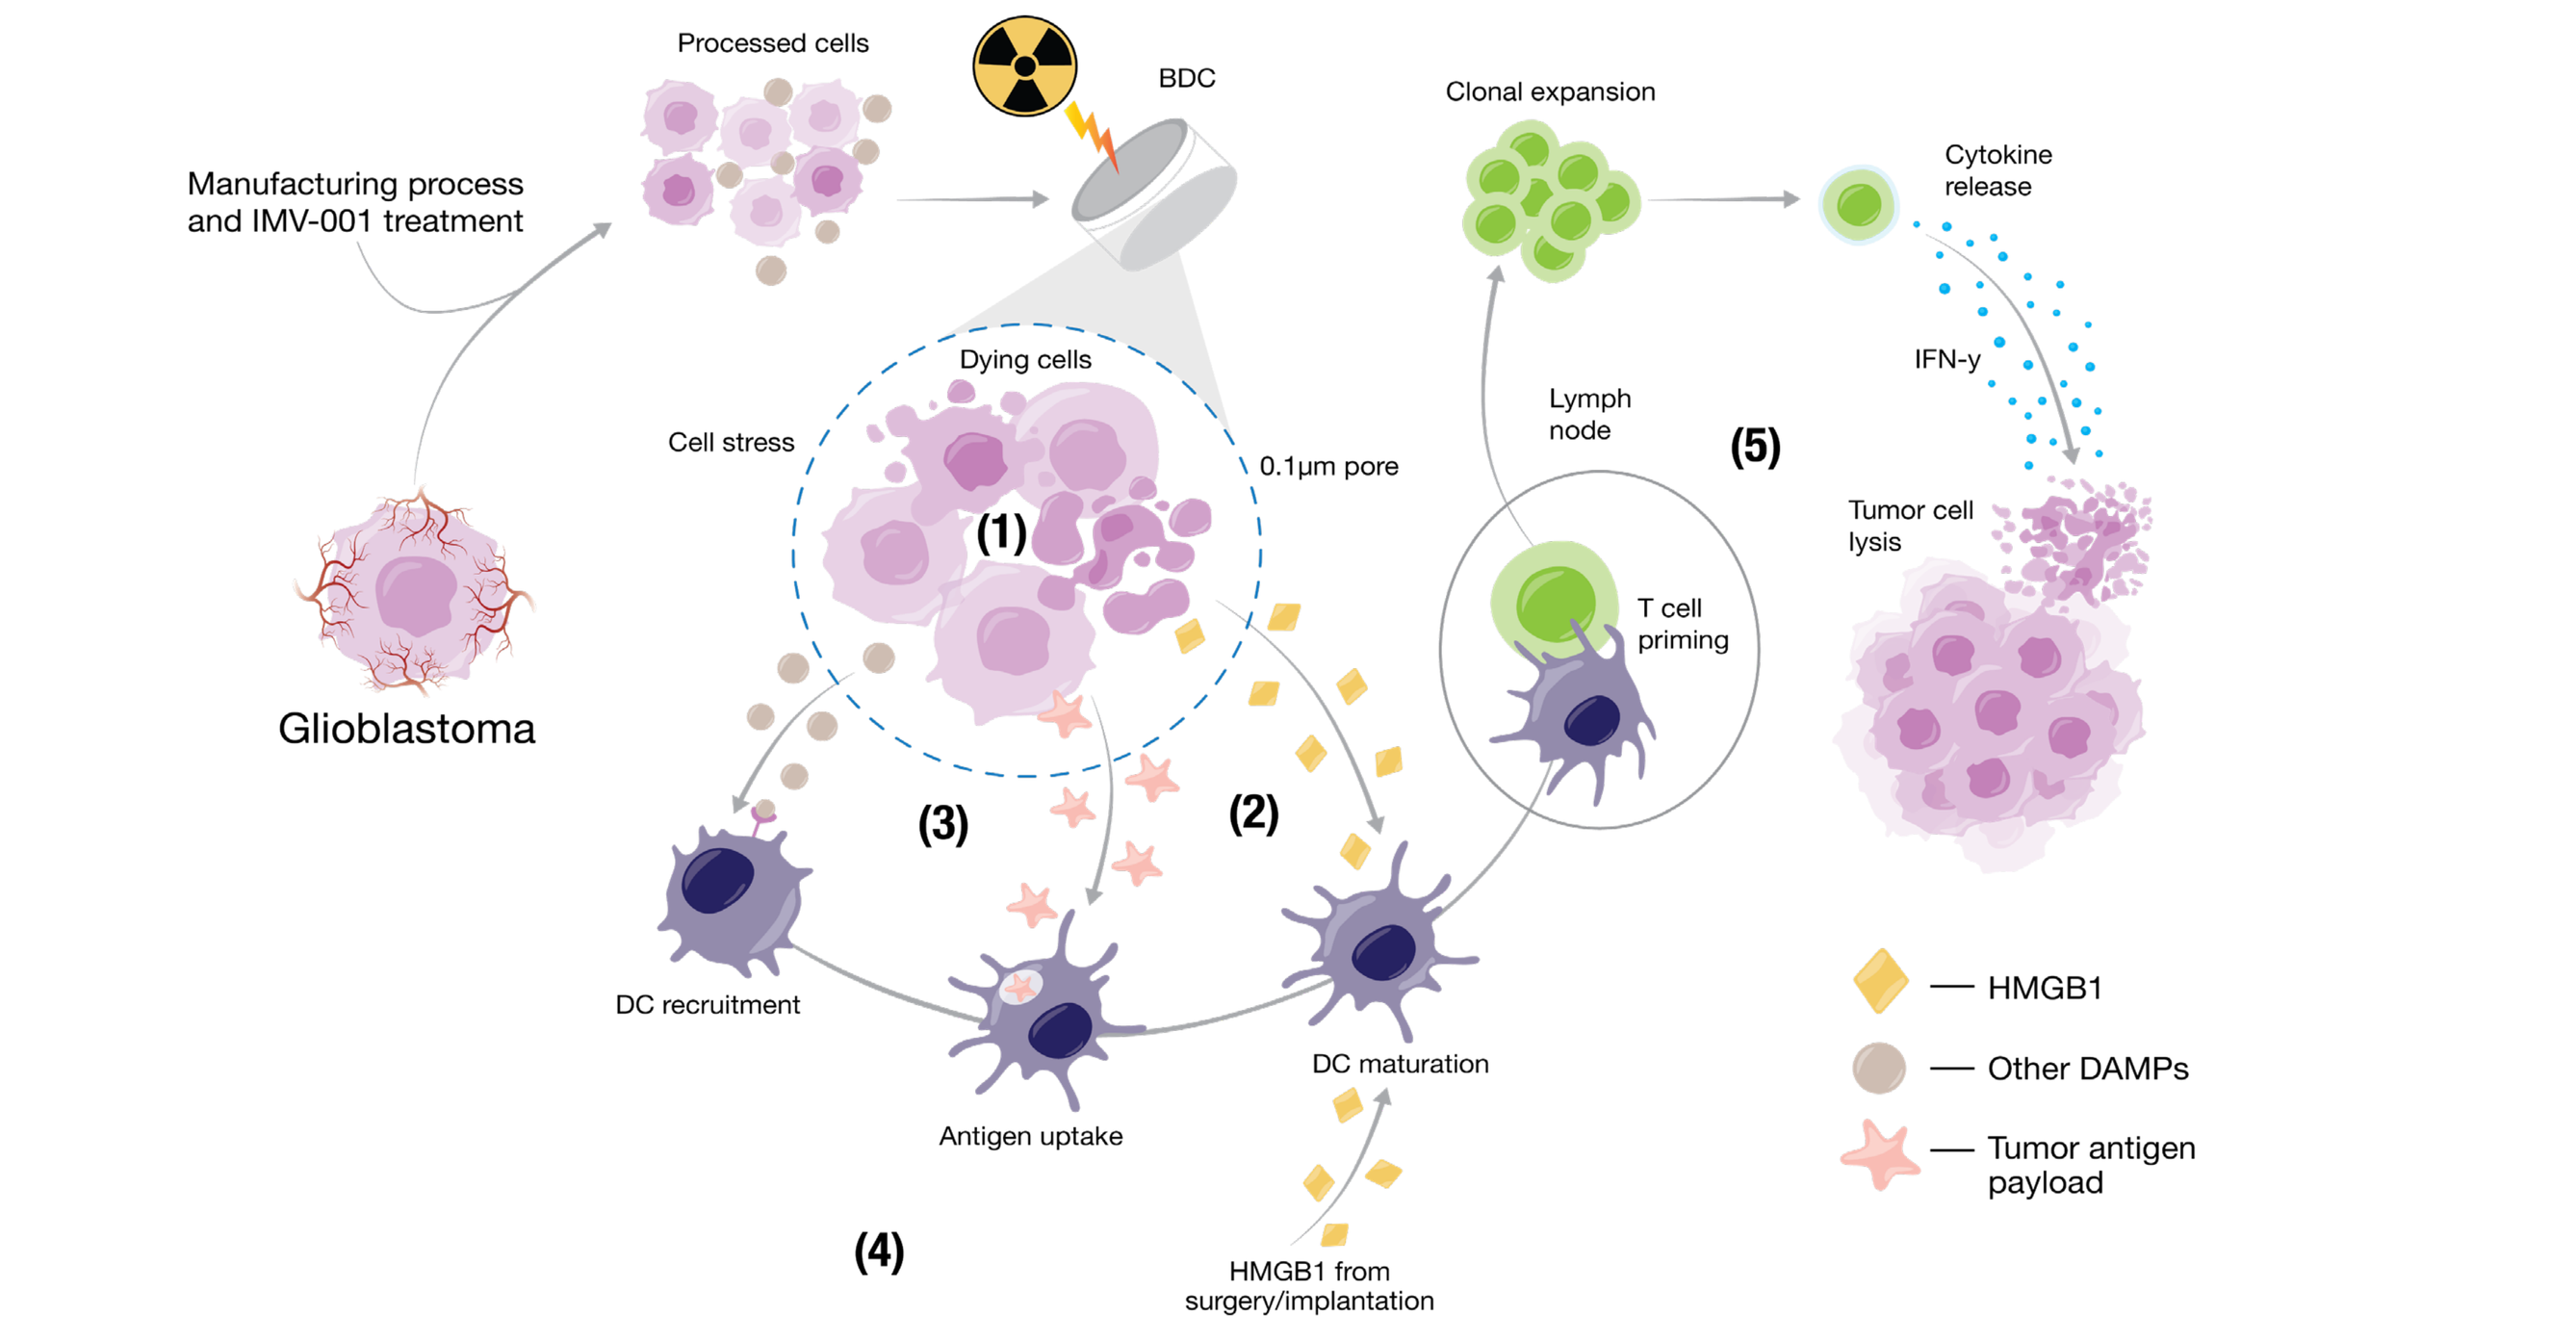

Manufacturing process and IMV-001 treatment
Processed cells
BDC
Clonal expansion
Cytokine release
IFN-γ
Glioblastoma
Cell stress
Dying cells
(1)
0.1µm pore
Lymph node
(5)
T cell priming
Tumor cell lysis
(3)
(2)
DC recruitment
Antigen uptake
DC maturation
(4)
HMGB1 from surgery/implantation
HMGB1
Other DAMPs
Tumor antigen payload

Processed cells removed at the time of glioblastoma resection are treated with IMV-001 (4 µg per chamber or 80 µg per dose). The combination drug product (IMV-001–treated processed autologous glioblastoma cells plus additional IMV-001) is filled into biodiffusion chambers (BDC), which are then irradiated for implantation into the abdomen of the patient. After implantation, the following stages occur: (1) processed cells undergo cellular stresses that result in cell death owing to irradiation, IMV-001 treatment, a low-nutrient environment, and an inability to adhere inside the BDC; (2) immunostimulatory factors (high mobility group box 1 [HMGB1] and damage-associated molecular patterns [DAMP] are released from stressed/dying cells inside the BDCs and mature local antigen presenting/dendritic cells (DC); (3) the antigens from the dying tumor cells diffuse out of the BDCs (<0.1 µm in size) and are taken up by the antigen presenting DC, while IMV-001 can inhibit immunosuppressive cells; (4) antigen-presenting cells migrate to lymph nodes; (5) where they activate tumor-specific T cells, which proliferate and undergo clonal expansion to promote a long-term adaptive immune response, and tumor antigen–specific T cells kill tumor cells remaining after surgery.

- Evidence of immune activation has been observed in preclinical experiments[15,16] and correlative clinical studies[13]
 - Dendritic cell maturation, CD4+ and CD8+ T-cell activation, and increase in central and effector memory T cells were observed in response to IGV-001 in vitro[13,15,16]
 - IGV-001 contributes to the induction of tumor immunity through multiple mechanisms, including the enhancement of antigen production by autologous tumor cells, inhibition of anti-inflammatory mechanisms, and the stimulation of antigen presentation in the patient (**Figure 2**)[17-19]
- In a phase 1b study (NCT02507583),[13] median PFS and OS compared favorably with SOC arms of published studies (**Figure 3**)[20-22]

Figure 3. Summary of phase 1b study data

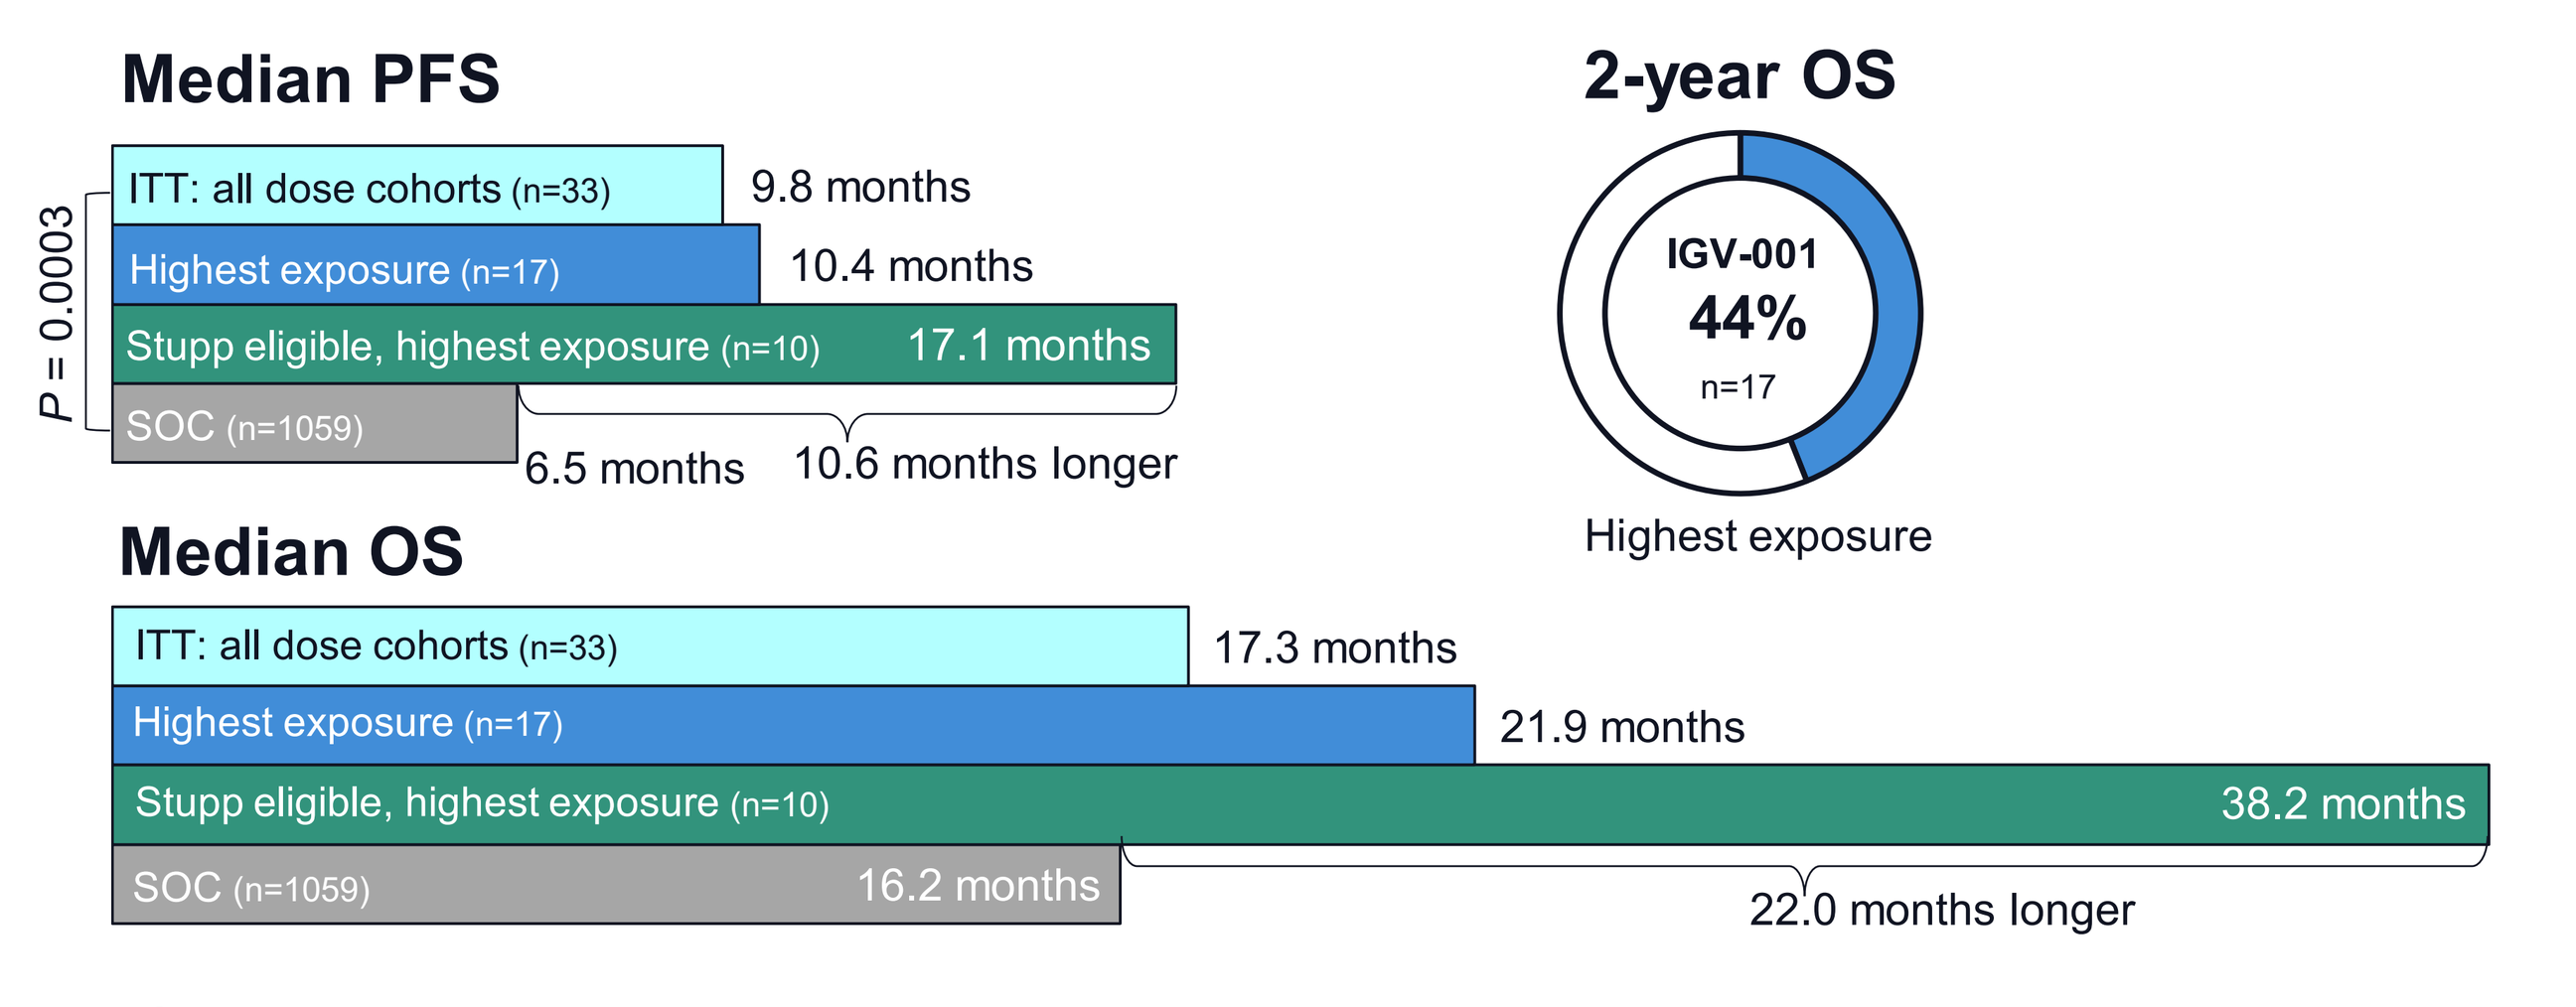

Median PFS
P = 0.0003
ITT: all dose cohorts (n=33)
9.8 months
Highest exposure (n=17)
10.4 months
Stupp eligible, highest exposure (n=10)
17.1 months
SOC (n=1059)
6.5 months
10.6 months longer
2-year OS
IGV-001
44%
n=17
Highest exposure
Median OS
ITT: all dose cohorts (n=33)
17.3 months
Highest exposure (n=17)
21.9 months
Stupp eligible, highest exposure (n=10)
38.2 months
SOC (n=1059)
16.2 months
22.0 months longer

IGV-001 was well tolerated and showed an exposure-response relationship, supporting the use, in subsequent clinical studies, of the highest exposure evaluated in the phase 1b study[13]

- Here, we describe the design and rationale of a randomized phase 2b study (NCT04485949) evaluating IGV-001 compared with placebo, both followed by SOC treatment in patients with newly diagnosed GBM

METHODS

Study objectives and study design

- The IGV-001 study (NCT04485949) is a multicenter, randomized, double-blind, placebo-controlled phase 2b study investigating the safety and efficacy of IGV-001 plus SOC (RT and TMZ treatments) versus placebo plus SOC in patients with newly diagnosed GBM (**Figure 4**)
- Resected GBM cancer cells treated with IMV-001 are encapsulated in biodiffusion chambers (BDCs) of 0.1 µm pore size, which allow tumor antigens and immune-stimulating molecules but not tumor cells to diffuse, then irradiated, producing IGV-001, which is implanted into 2 abdominal sites (between the rectus abdominis muscle and fascia) of patients for 48 to 52 hours, then explanted (**Figure 1**)
- Patients will be randomized 2:1 to either receive IGV-001 at 16-20 BDCs or placebo for 48 to 52 hours and stratified by age groups (≤50 years vs >50 years at randomization)
 - The BDCs implanted in patients in the placebo group contain inactive solution without GBM tumor cells and without IMV-001
- Six weeks after randomization, patients will receive RT (54-60 Gy total dose delivered as 2 Gy per fraction) per institutional standards (hence per investigators' choice) for 5 days per week along with TMZ (75 mg/m^2 orally) once daily for 6 weeks
- Four weeks after completion of RT, patients will receive TMZ maintenance (150-200 mg/m^2 orally) on days 1-5 of each 28-day cycle for 6 cycles (week 41)

Figure 4. Study design

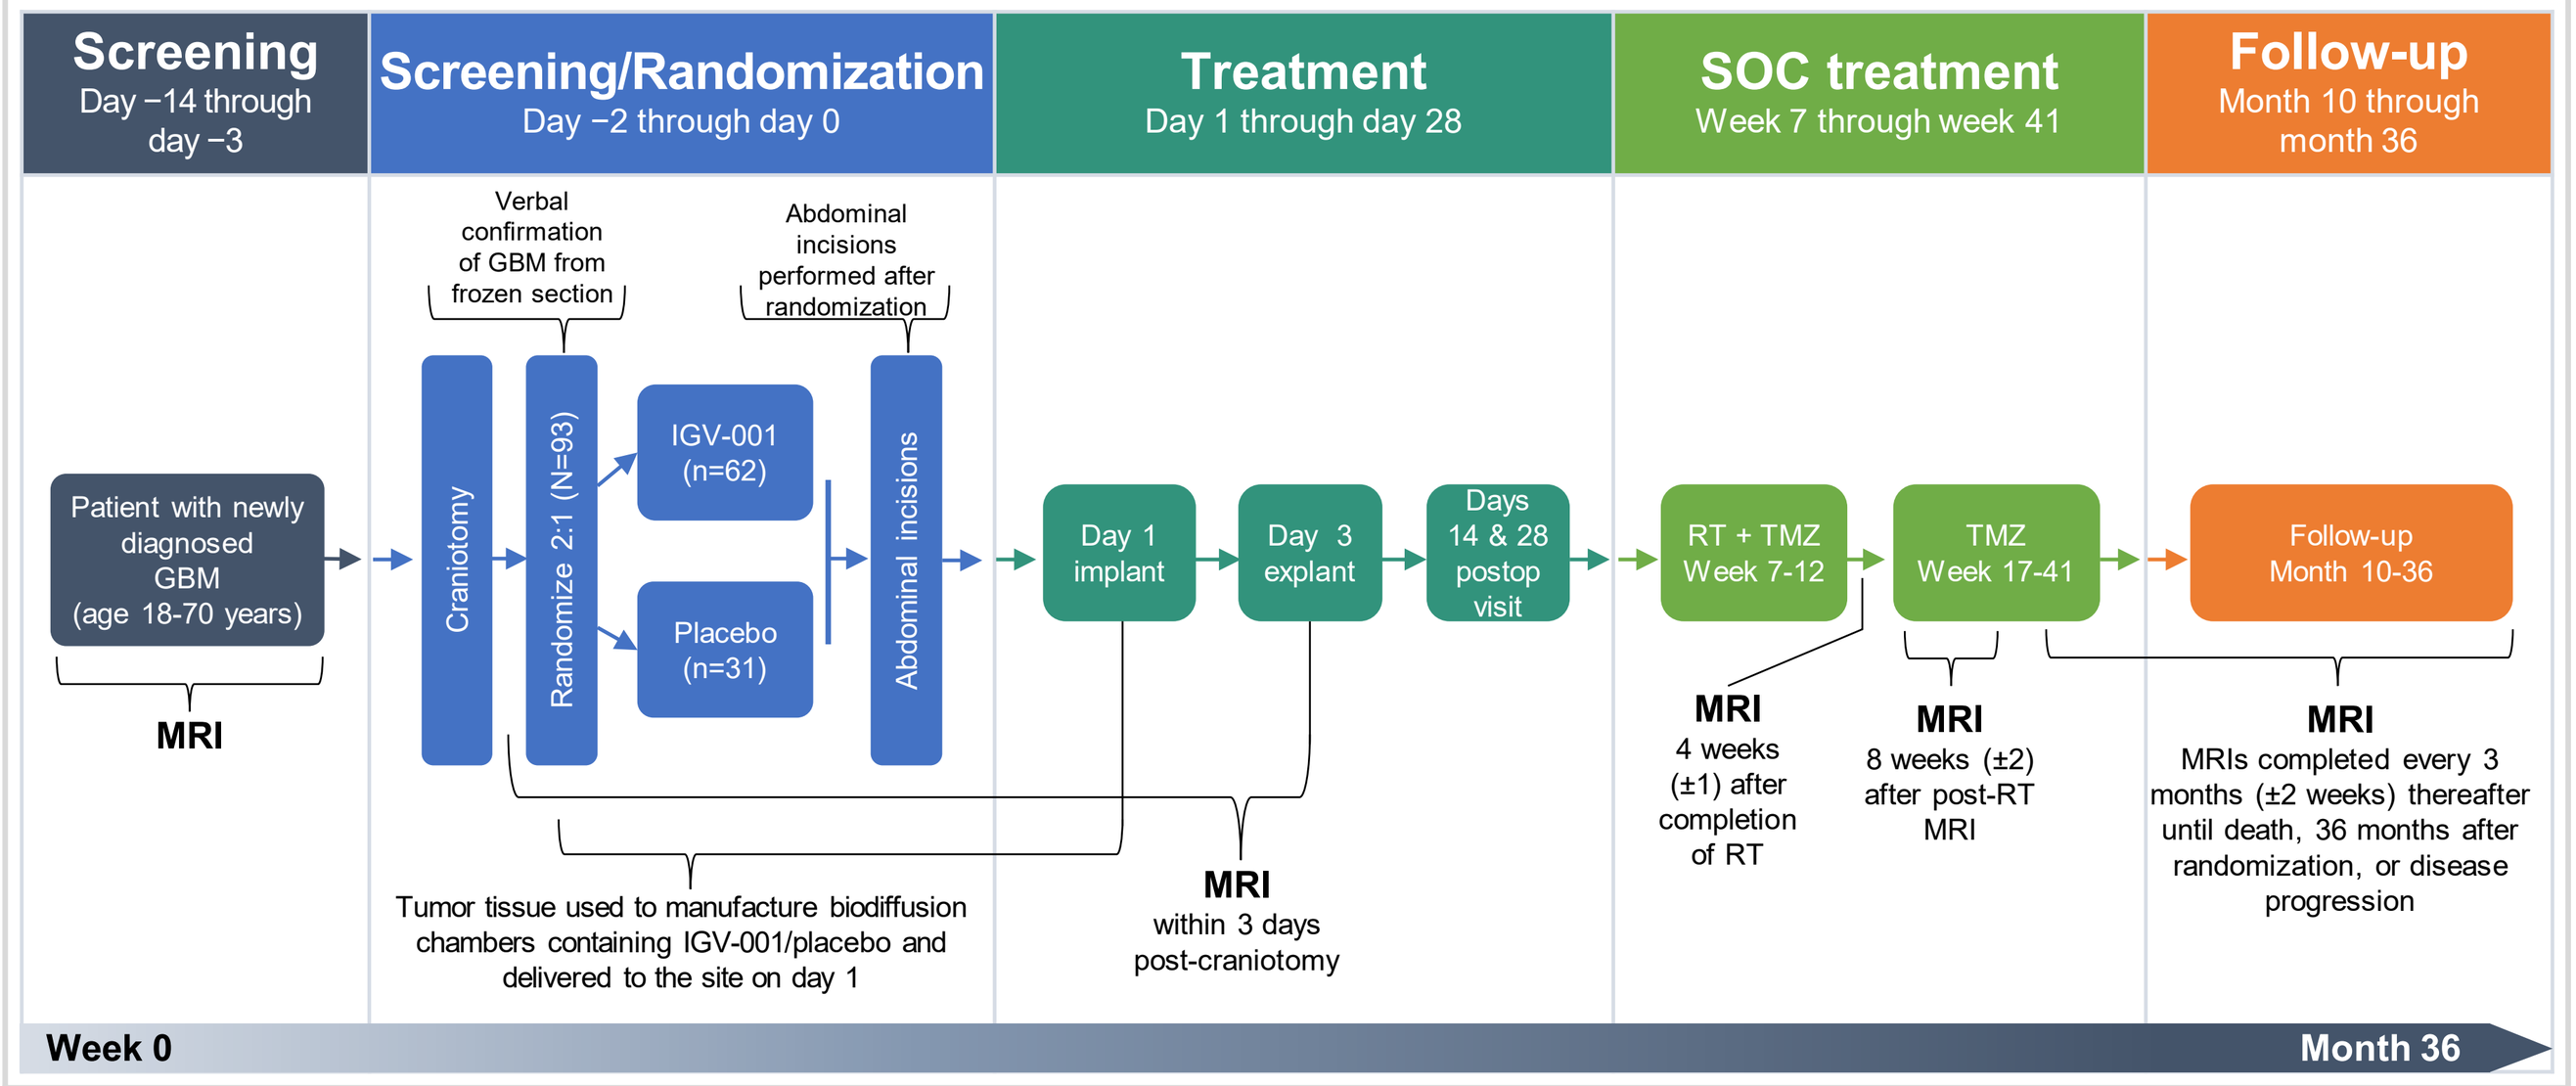

Screening
Day −14 through day −3
Screening/Randomization
Day −2 through day 0
Treatment
Day 1 through day 28
SOC treatment
Week 7 through week 41
Follow-up
Month 10 through month 36
Verbal confirmation of GBM from frozen section
Abdominal incisions performed after randomization
Patient with newly diagnosed GBM (age 18-70 years)
MRI
Craniotomy
Randomize 2:1 (N=93)
IGV-001 (n=62)
Placebo (n=31)
Abdominal incisions
Day 1 implant
Day 3 explant
Days 14 & 28 postop visit
RT + TMZ Week 7-12
TMZ Week 17-41
Follow-up Month 10-36
MRI
4 weeks (±1) after completion of RT
MRI
8 weeks (±2) after post-RT MRI
MRI
MRIs completed every 3 months (±2 weeks) thereafter until death, 36 months after randomization, or disease progression
Tumor tissue used to manufacture biodiffusion chambers containing IGV-001/placebo and delivered to the site on day 1
MRI
within 3 days post-craniotomy
Week 0
Month 36

GBM, glioblastoma; MRI, magnetic resonance imaging; RT, radiotherapy; SOC, standard of care; TMZ, temozolomide.

Table 1. Primary and secondary study endpoints

Primary endpoint	Secondary endpoint	Tertiary endpoints	Exploratory endpoints	Safety endpoint
PFS in patients with newly diagnosed GBM treated with IGV-001 versus patients treated with placebo	OS in patients treated with IGV-001 versus patients treated with placebo	• Time to deterioration of KPS score • PFS • OS within subgroups of patients with methylated MGMT+ and unmethylated MGMT− • PFS, OS within the subgroup of patients with histologic confirmation of WHO Grade 3 (diffuse astrocytic glioma, IDH wild type, with molecular features of WHO Grade 4 GBM) or WHO Grade 4 GBM	• QOL • Immune response markers • Response rate in patients who have measurable residual disease after surgery • Tumor mutational burden	Determine safety and tolerability of IGV-001 in patients with newly diagnosed GBM

GBM, glioblastoma; IDH, isocitrate dehydrogenase; KPS, Karnofsky Performance Status; MGMT, methylguanine-DNA methyltransferase; OS, overall survival; PFS, progression-free survival; QOL, quality of life; WHO, World Health Organization.

Key inclusion criteria

- Adult aged ≥18 and ≤70 years at screening
- Karnofsky Performance Status (KPS) score ≥70 at screening
- Diagnosis of GBM (histologic and/or WHO Grade 4 molecular diffuse astrocytoma) with confirmation from intraoperative frozen section
- Diagnostic contrast-enhanced magnetic resonance imaging (MRI) scan with fluid-attenuated inversion recovery sequence of the brain and thin cuts (1-1.5 mm) at screening. Patients must have a resectable contrast-enhancing lesion preoperatively with a total biperpendicular product of 4 cm^2 in 2 different planes (axial, sagittal, or coronal)
- Tumor location in the supratentorial compartment
- Acceptable laboratory parameters and adequate bone marrow and organ function

Key exclusion criteria

- Bihemispheric disease, multicentric disease, or disease burden involving the brainstem or cerebellum based on MRI after gadolinium enhancement
- Any previous surgical resection or any anticancer intervention for GBM
- Recurrent glioma, a concurrent malignancy, or malignancy within 3 years of randomization

Study endpoints

- Endpoints are summarized in **Table 1**
- The primary outcome is PFS, defined as the time from randomization to event or censoring, as determined by blinded central radiology review
- Secondary outcomes include OS, defined as the time from randomization to death due to any cause, and safety

Statistical methods

- The intention-to-treat (ITT) analysis and safety analysis sets are defined as all randomized patients
- The analysis of the primary endpoint PFS will be triggered when ≥55 PFS events in the ITT analysis set have been observed per blinded central radiology review, based on Response Assessment in Neuro-Oncology (RANO) criteria,[23] and will be performed using a stratified log-rank test and a 1-sided 0.05 significance level
- The study is designed to achieve 80% power at a 1-sided α of 0.05 to detect a statistically significant difference in PFS between groups
 - Assuming an accrual period of ~7 months, a 7% yearly rate for loss to imaging follow-up, and 2:1 randomization, approximately 93 patients will be randomized to observe 55 PFS events in the ITT analysis set by 11-12 months after the last patient is randomized
- Approximately 36 months after randomization of the last patient, the final analysis of OS will be performed using a stratified log-rank test and a 1-sided 0.05 significance level, adjusted using the Benjamini-Hochberg approach[24]
- Tertiary efficacy objectives will have multivariate analyses performed using the Cox proportional hazards model for PFS and OS to determine independent prognostic factors
 - The covariates evaluated for the multivariate models will be the assigned treatment group; age group; MGMT methylation status (MGMT+ or MGMT−); histologic confirmation of either WHO Grade 3, WHO Grade 4 GBM, or diffuse astrocytoma; IDH-mutated, with any histologic feature of GBM and/or a *CDKN2A/B* mutation; and extent of resection (gross, subtotal, or partial)
- KPS score will be analyzed over time using descriptive statistics. Time to deterioration of the KPS score will be analyzed using the product-limit method
- Safety will be reported as the incidence of procedure-related adverse events and treatment-emergent adverse events from the time of randomization and will be graded according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 5.0
 - Safety data will be reported overall as well as separately for the screening period, the treatment period, and the SOC treatment period until the 30-day safety visit

STATUS

- The study is ongoing and is planned for approximately 25 centers in the United States (**Figure 5**)
- As of July 7th , there are 18 open sites

Figure 5. Anticipated study locations

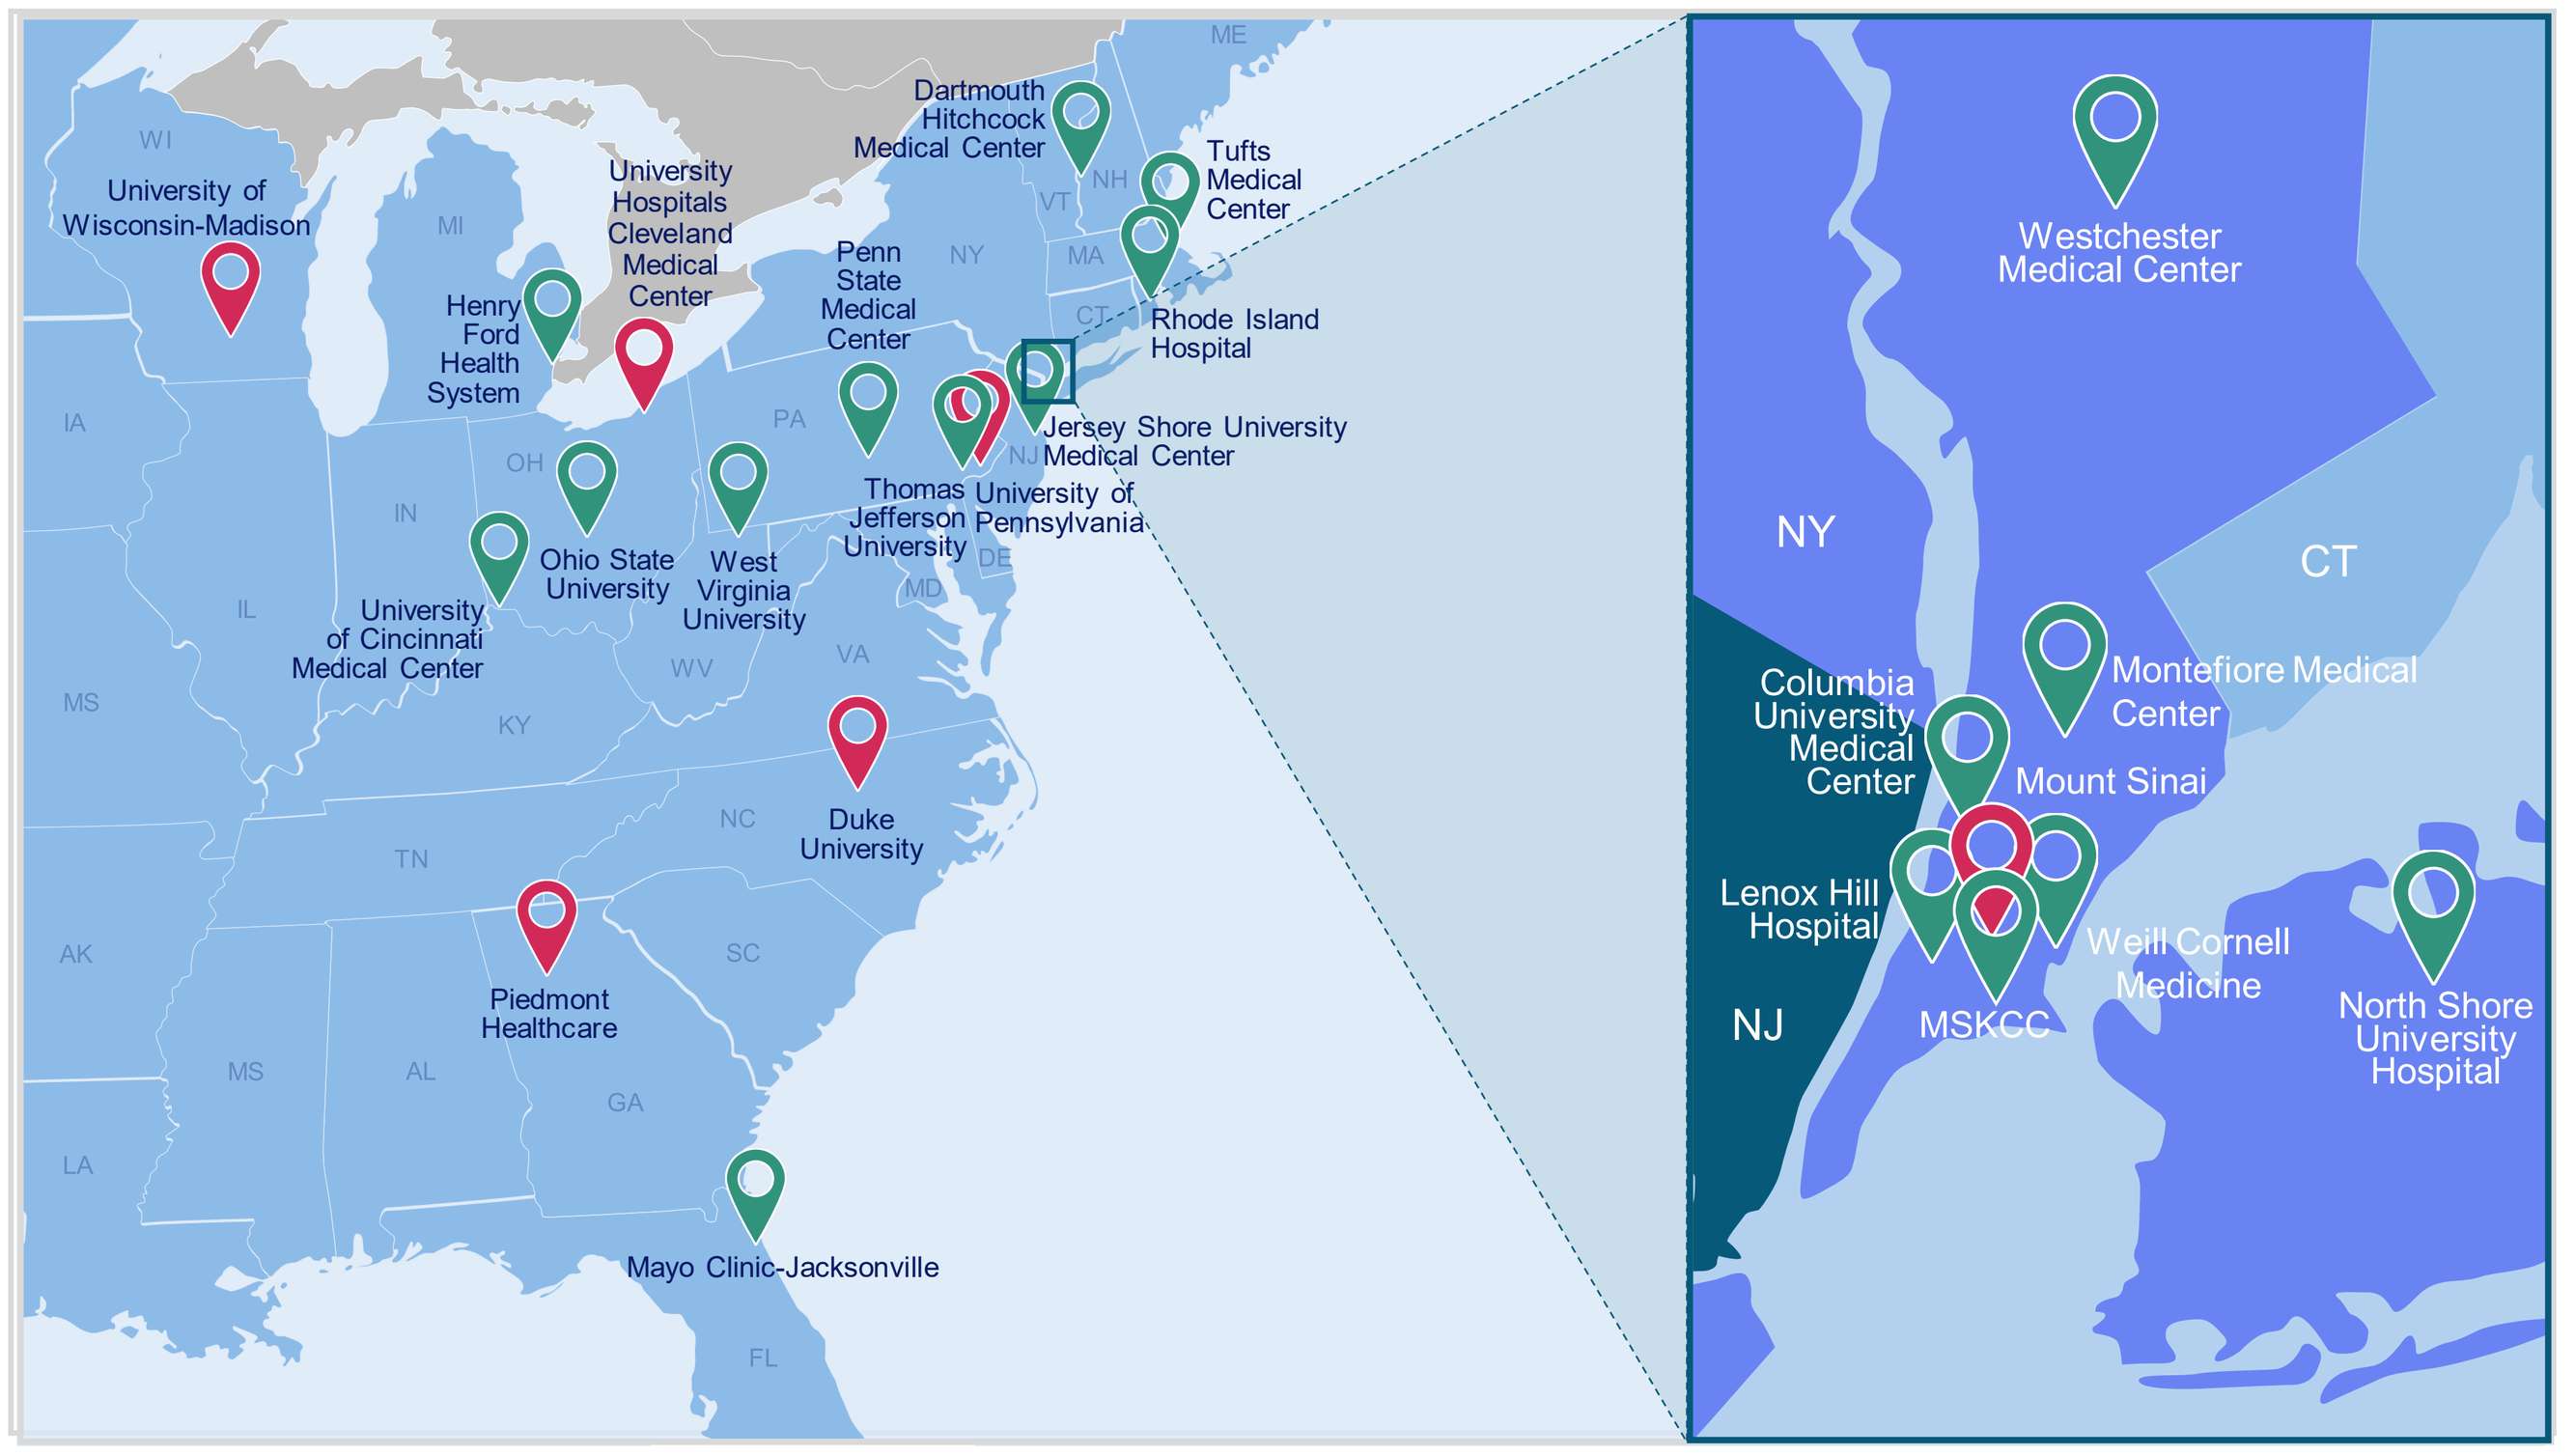

University of Wisconsin-Madison
Henry Ford Health System
University Hospitals Cleveland Medical Center
Penn State Medical Center
Dartmouth Hitchcock Medical Center
Tufts Medical Center
Rhode Island Hospital
Jersey Shore University Medical Center
Thomas Jefferson University
University of Pennsylvania
Ohio State University
West Virginia University
University of Cincinnati Medical Center
Duke University
Piedmont Healthcare
Mayo Clinic-Jacksonville
Westchester Medical Center
NY
CT
NJ
Columbia University Medical Center
Montefiore Medical Center
Mount Sinai
Lenox Hill Hospital
Weill Cornell Medicine
MSKCC
North Shore University Hospital

ACKNOWLEDGMENTS

Funding for the IGV-001 study and medical writing support were provided by Imvax, Inc. (Philadelphia, PA, USA). Figure 2 was created with BioRender.com. Medical writing was provided by Emily Cullinan, PhD, CMPP, and Francesca Balordi, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA).

REFERENCES

1. Stupp R, et al. *N Engl J Med.* 2005;352(10):987-996. **2.** Maris C, et al. *Br J Cancer.* 2015;113(5):729-737. **3.** Larsson O, et al. *Br J Cancer.* 2005;92(12):2097-2101. **4.** Philippou A, et al. *Front Endocrinol (Lausanne).* 2013;4:31. **5.** Philippou A, et al. *Mutat Res Rev Mutat Res.* 2017;772:105-122. **6.** Scartozzi M, et al. *Discov Med.* 2011;11(57):144-153. **7.** Dunn SE, et al. *Cancer Res.* 1998;58(15):3353-3361. **8.** Nakamura S, et al. *Exp Cell Res.* 1997;235(1):287-294. **9.** Turner BC, et al. *Cancer Res.* 1997;57(15):3079-3083. **10.** Exley MA, et al. *J Clin Med.* 2022;11(4):1069. **11.** Ryan PD, et al. *Oncologist.* 2008;13(1):16-24. **12.** Sachdev D, et al. *Mol Cancer Ther.* 2007;6(1):1-12. **13.** Andrews DW, et al. *Clin Cancer Res.* 2021;27(7):1912-1922. **14.** Andrews DW, et al. *J Clin Oncol.* 2001;19(8):2189-2200. **15.** Zellander A, et al. *J Immunother Cancer.* 2022;10(suppl 2):A1113 (Abstract 1398). **16.** Zilberberg J, et al. *Cancer Res.* 2022;82(suppl 12):Abstract 626. **17.** Cultrara C, et al. *J Immunother Cancer.* 2022;10(suppl 2):A1113 (Abstract 1071). **18.** Uhl C, et al. CRI-ENCI-AACR Sixth International Cancer Immunotherapy Conference (CICON) 2022; Abstract A001. **19.** Zilberberg J, et al. *J Immunother Cancer.* 2021;9(suppl 2):A231 (Abstract 218). **20.** Chinot OL, et al. *N Engl J Med.* 2014;370(8):709-722. **21.** Gilbert MR, et al. *N Engl J Med.* 2014;370(8):699-708. **22.** Stupp R, et al. *Lancet Oncol.* 2009;10(5):459-466. **23.** Wen PY, et al. *J Clin Oncol.* 2017;35(21):2439-2449. **24.** Benjamini Y, et al. *J R Stat Soc Series B Stat Methodol.* 1995;57(1):289-300.

Presented at SNO/ASCO CNS Cancer Conference (SNO/ASCO) | August 10-12, 2023 | San Francisco, CA, USA. © Copyright 2023 IMVAX®, Inc. All rights reserved.