ROS-Dependent Activation of Immunogenic Glioblastoma Cell Death and Release of Immunogenic Particles by an Autologous Cell-Based Immunotherapeutic Platform

Christopher Cultrara,[1] Kenneth Kirby,[1] Essam Elrazaq,[1] Christopher Uhl,[1] Amelia Zellander,[1] Lorenzo Galluzzi,[2] Mark A. Exley,[1] Jenny Zilberberg[1]

[1]Imvax, Inc.; [2]Department of Radiation Oncology, Weill Cornell Medical College

#1812

BACKGROUND

Imvax is developing a novel personalized immunotherapy platform that combines whole-tumor derived cells with an antisense oligonucleotide against the insulin-like growth factor 1-receptor in implantable biodiffusion chambers (BDCs; 0.1 mm pore size). The lead product, IGV-001, was evaluated in newly diagnosed glioblastoma (GBM) patients in a Phase 1b clinical trial.[1] Median overall survival of the highest exposure IGV-001-treated "Stupp-eligible"[2] patients (n = 10) was 38.2 months compared with 16.2 months in current standard-of-care-treated patients (p = 0.044)[1] [NCT02507583]. Imvax also reported anti-tumor activity of the murine variant of this product, *m*IGV-001, in luciferase-expressing[3] and wild-type GL261 mouse GBM cells, detecting *m*IGV-001-induced immune responses in BDC-draining lymph nodes.[4] Since reactive oxygen species (ROS) overproduction can result in immunogenic cell death,[5] we investigated the role of ROS formation as a mediator of cell death in *m*IGV-001, as well as the generation of subcellular particles that, when released from the BDCs, may provide a tumor antigen payload with potential anti-tumor activity.

METHODS

Mouse (*m*) or human (*h*) variants of IGV-001 were prepared[3,4] using mouse GL261 or human T98G GBM cells, respectively, and BDCs were incubated at 37°C and 5% CO_2 for 24-48 h. ROS levels were detected by flow cytometry and fluorescence microscopy via oxidation of H2DCF (a fluorescein analog used to quantify ROS in cells) in the presence or absence of the antioxidant N-acetyl-cysteine (NAC). *m/h*IGV-001 viability was assessed by flow cytometry using Annexin V/7-AAD (Biotium) staining. Particle size distribution in *h*IGV-001 BDC contents was analyzed using a Nanosight NS300 (Malvern Panalytical). The transport of subcellular particles (25-90 nm) across the BDC membrane was modeled under dynamic and static conditions using MATLAB® 2022a (Mathworks™) and confirmed by in vitro studies.

RESULTS

- IGV-001-draining lymph nodes are enriched in mature antigen-presenting cells and lymphoid effector cells (fig. 1).
- IGV-001 imposes oxidative and endoplasmic reticulum (ER) stress onto GBM cells (fig. 2).
- Particle analysis confirmed that during 48 h in vitro incubation, a human (*h*) variant of IGV-001 prepared with T98G human GBM cells produces particles that can efficiently diffuse through the BDC membrane (<0.1 µm; fig. 3).

FIGURE 1

IGV-001-draining lymph nodes are enriched in mature antigen-presenting cells and lymphoid effector cells

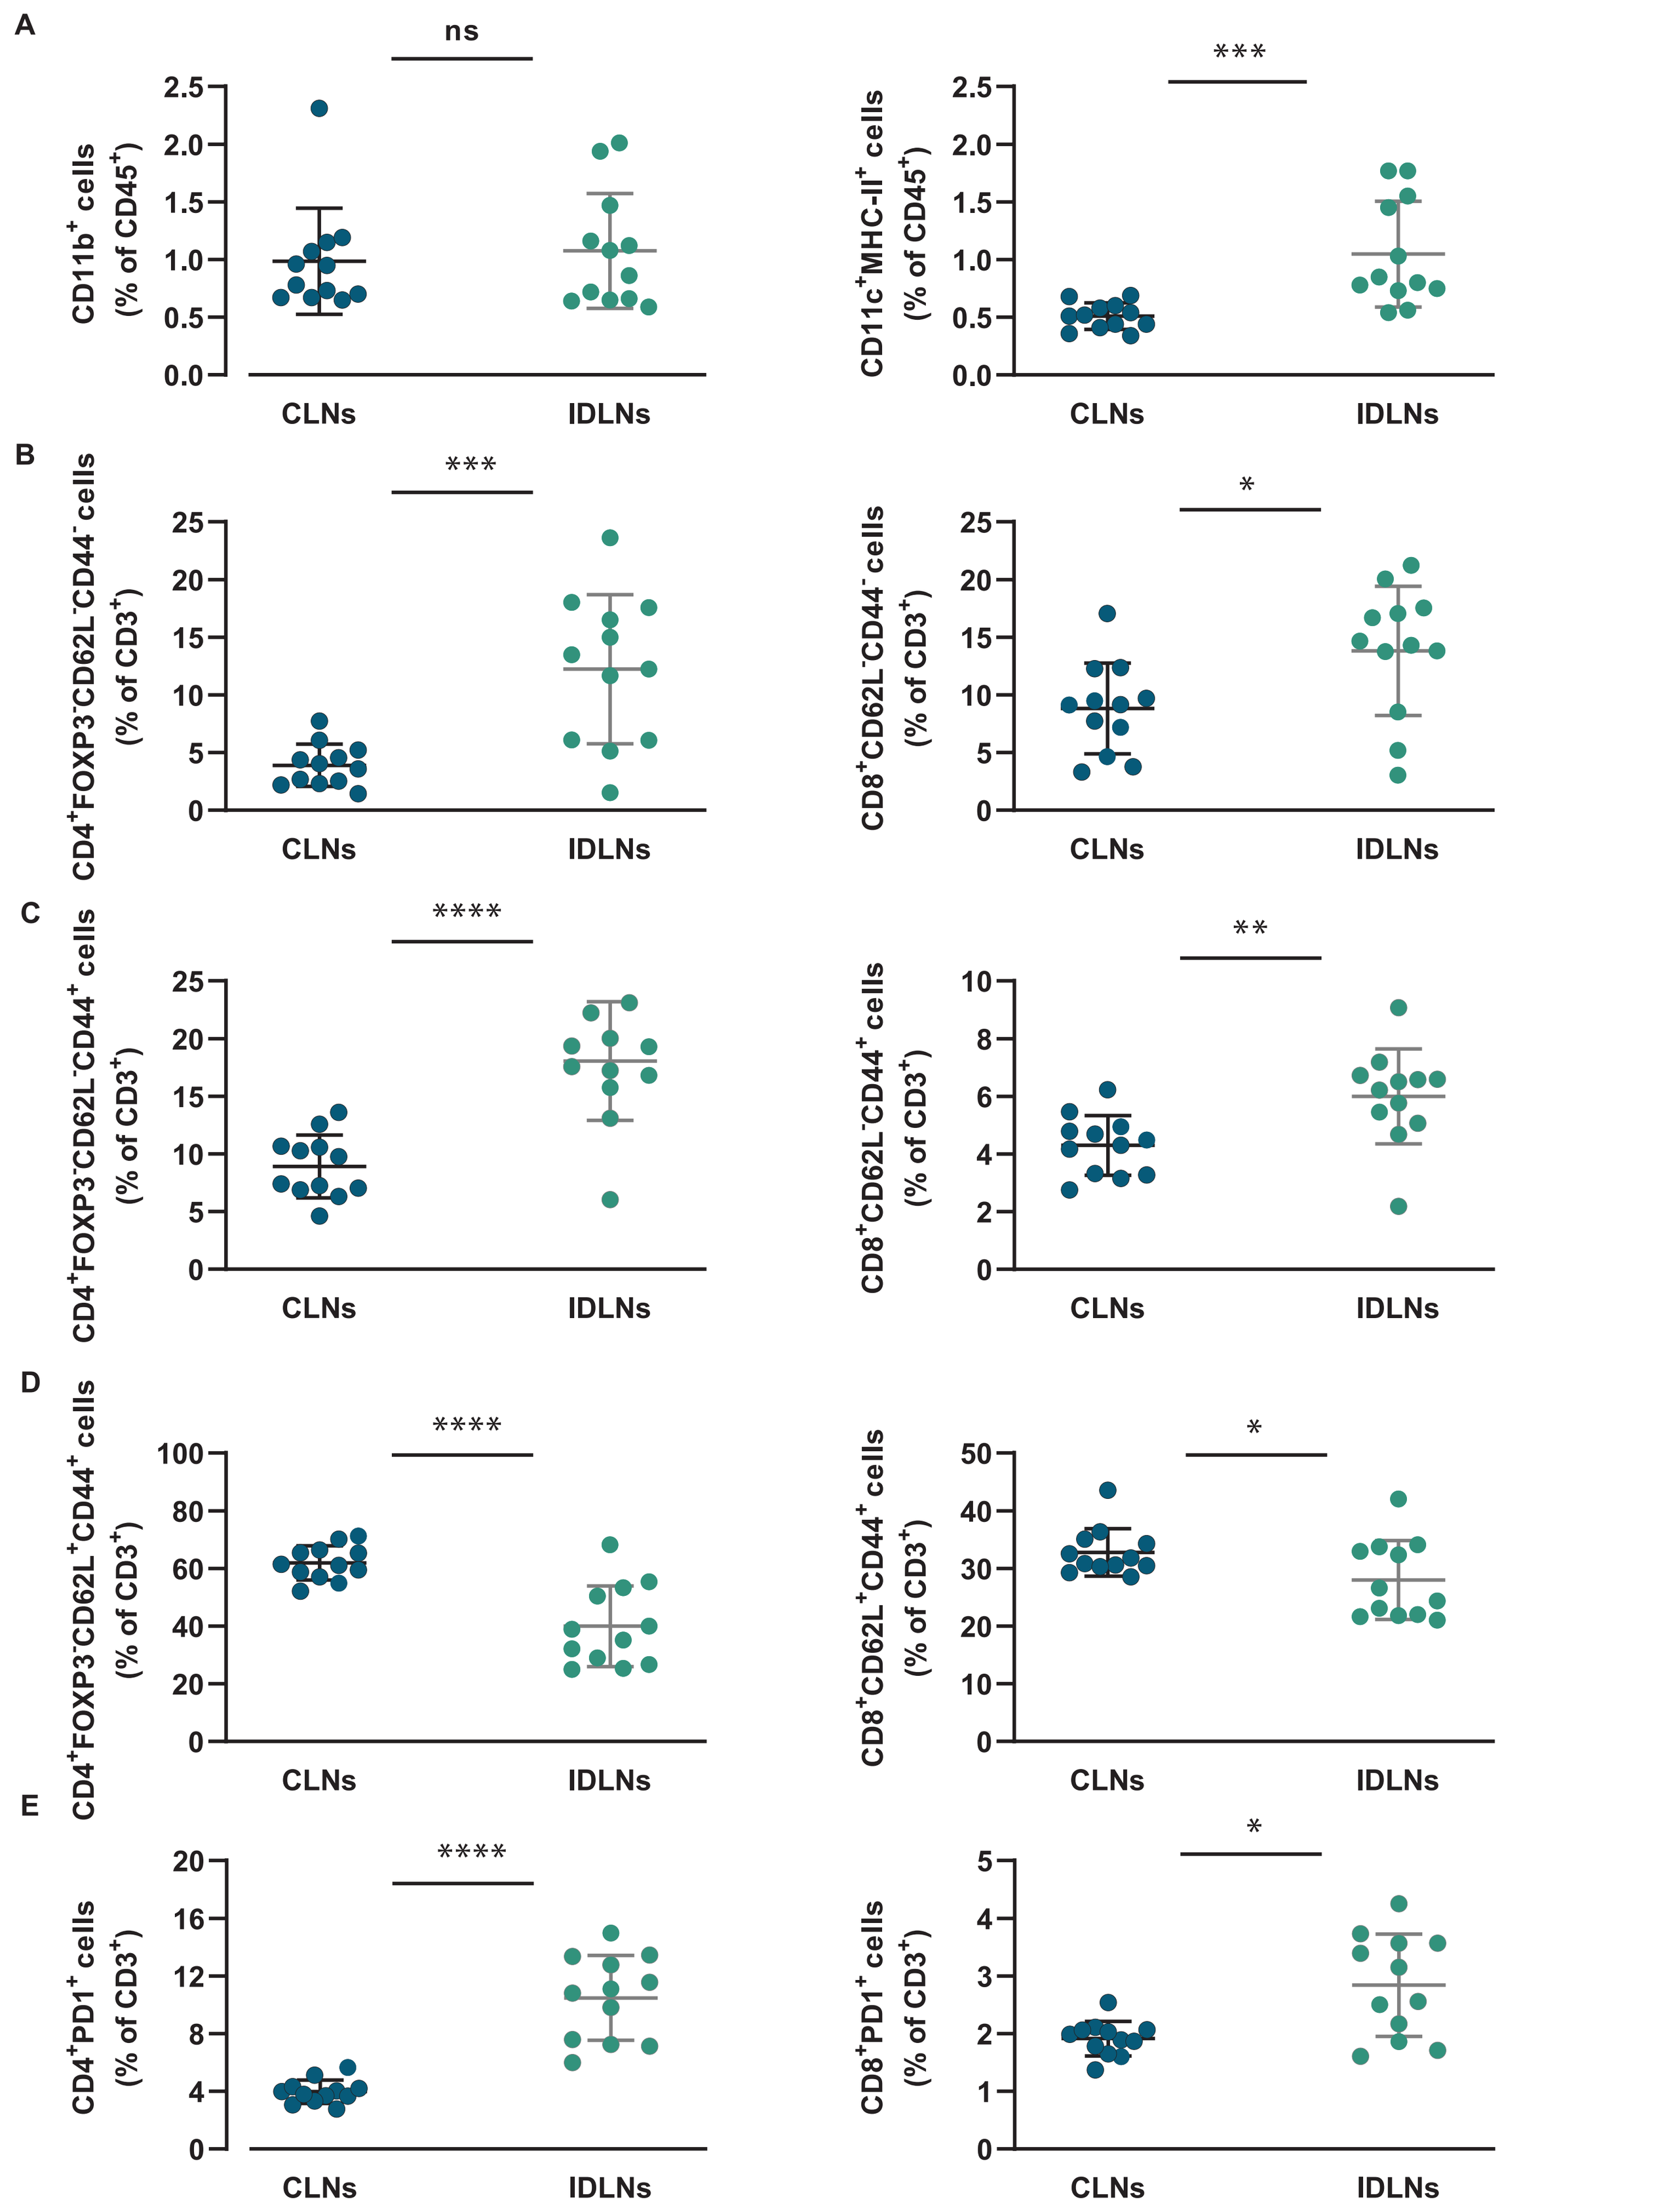
A
ns
CD11b+ cells (% of CD45+)

CD11c+MHC-II+ cells (% of CD45+)
B

CD4+FOXP3-CD62L-CD44- cells (% of CD3+)
*
CD8+CD62L-CD44- cells (% of CD3+)
C

CD4+FOXP3-CD62L-CD44+ cells (% of CD3+)
**
CD8+CD62L-CD44+ cells (% of CD3+)
D

CD4+FOXP3-CD62L+CD44+ cells (% of CD3+)
*
CD8+CD62L+CD44+ cells (% of CD3+)
E

CD4+PD1+ cells (% of CD3+)
*
CD8+PD1+ cells (% of CD3+)
CLNs
IDLNs

Figure 1. Mice were exposed to *m*IGV-001 (manufactured with 1×10[6] GL261-luc2 cells/BDC). BDCs were implanted in the flank and left in place for 48 h. A subcutaneous tumor challenge with GL261-luc2 (1×10[7] cells/0.2 mL) was performed 26 days after BDC explantation on the same flank, and mice were sacrificed 7 days later. Lymph nodes proximal to the *m*IGV-001 BDCs (IDLNs) and non draining lymph nodes in the contralateral site (CLNs) were isolated and analyzed by flow cytometry to compare the distribution of myeloid and T-cell subsets. Percentages are noted for the following: **(A)** $CD45^+CD11b^+$ myeloid cells and $CD45^+CD11c^+MHC\text{-}II^+$ dendritic cells, **(B)** $CD3^+CD4^+FOXP3^-CD62L^-CD44^-$ and $CD3^+CD8^+CD62L^-CD44^-$ effector T cells, **(C)** $CD3^+CD4^+FOXP3^-CD62L^-CD44^+$ and $CD3^+CD8^+CD62L^-CD44^+$ effector memory T cells, **(D)** $CD3^+CD4^+FOXP3^-CD62L^+CD44^+$ and $CD3^+CD8^+CD62L^+CD44^+$ central memory T cells, and **(E)** PD-1 expression on $CD4^+$ and $CD8^+$ T cells. Results are presented as mean ± standard deviation (SD) of n = 12 mice per group. Statistical analysis was performed using an unpaired t-test, *p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001.

FIGURE 2

IGV-001 imposes oxidative and endoplasmic reticulum stress on GBM cells in vitro

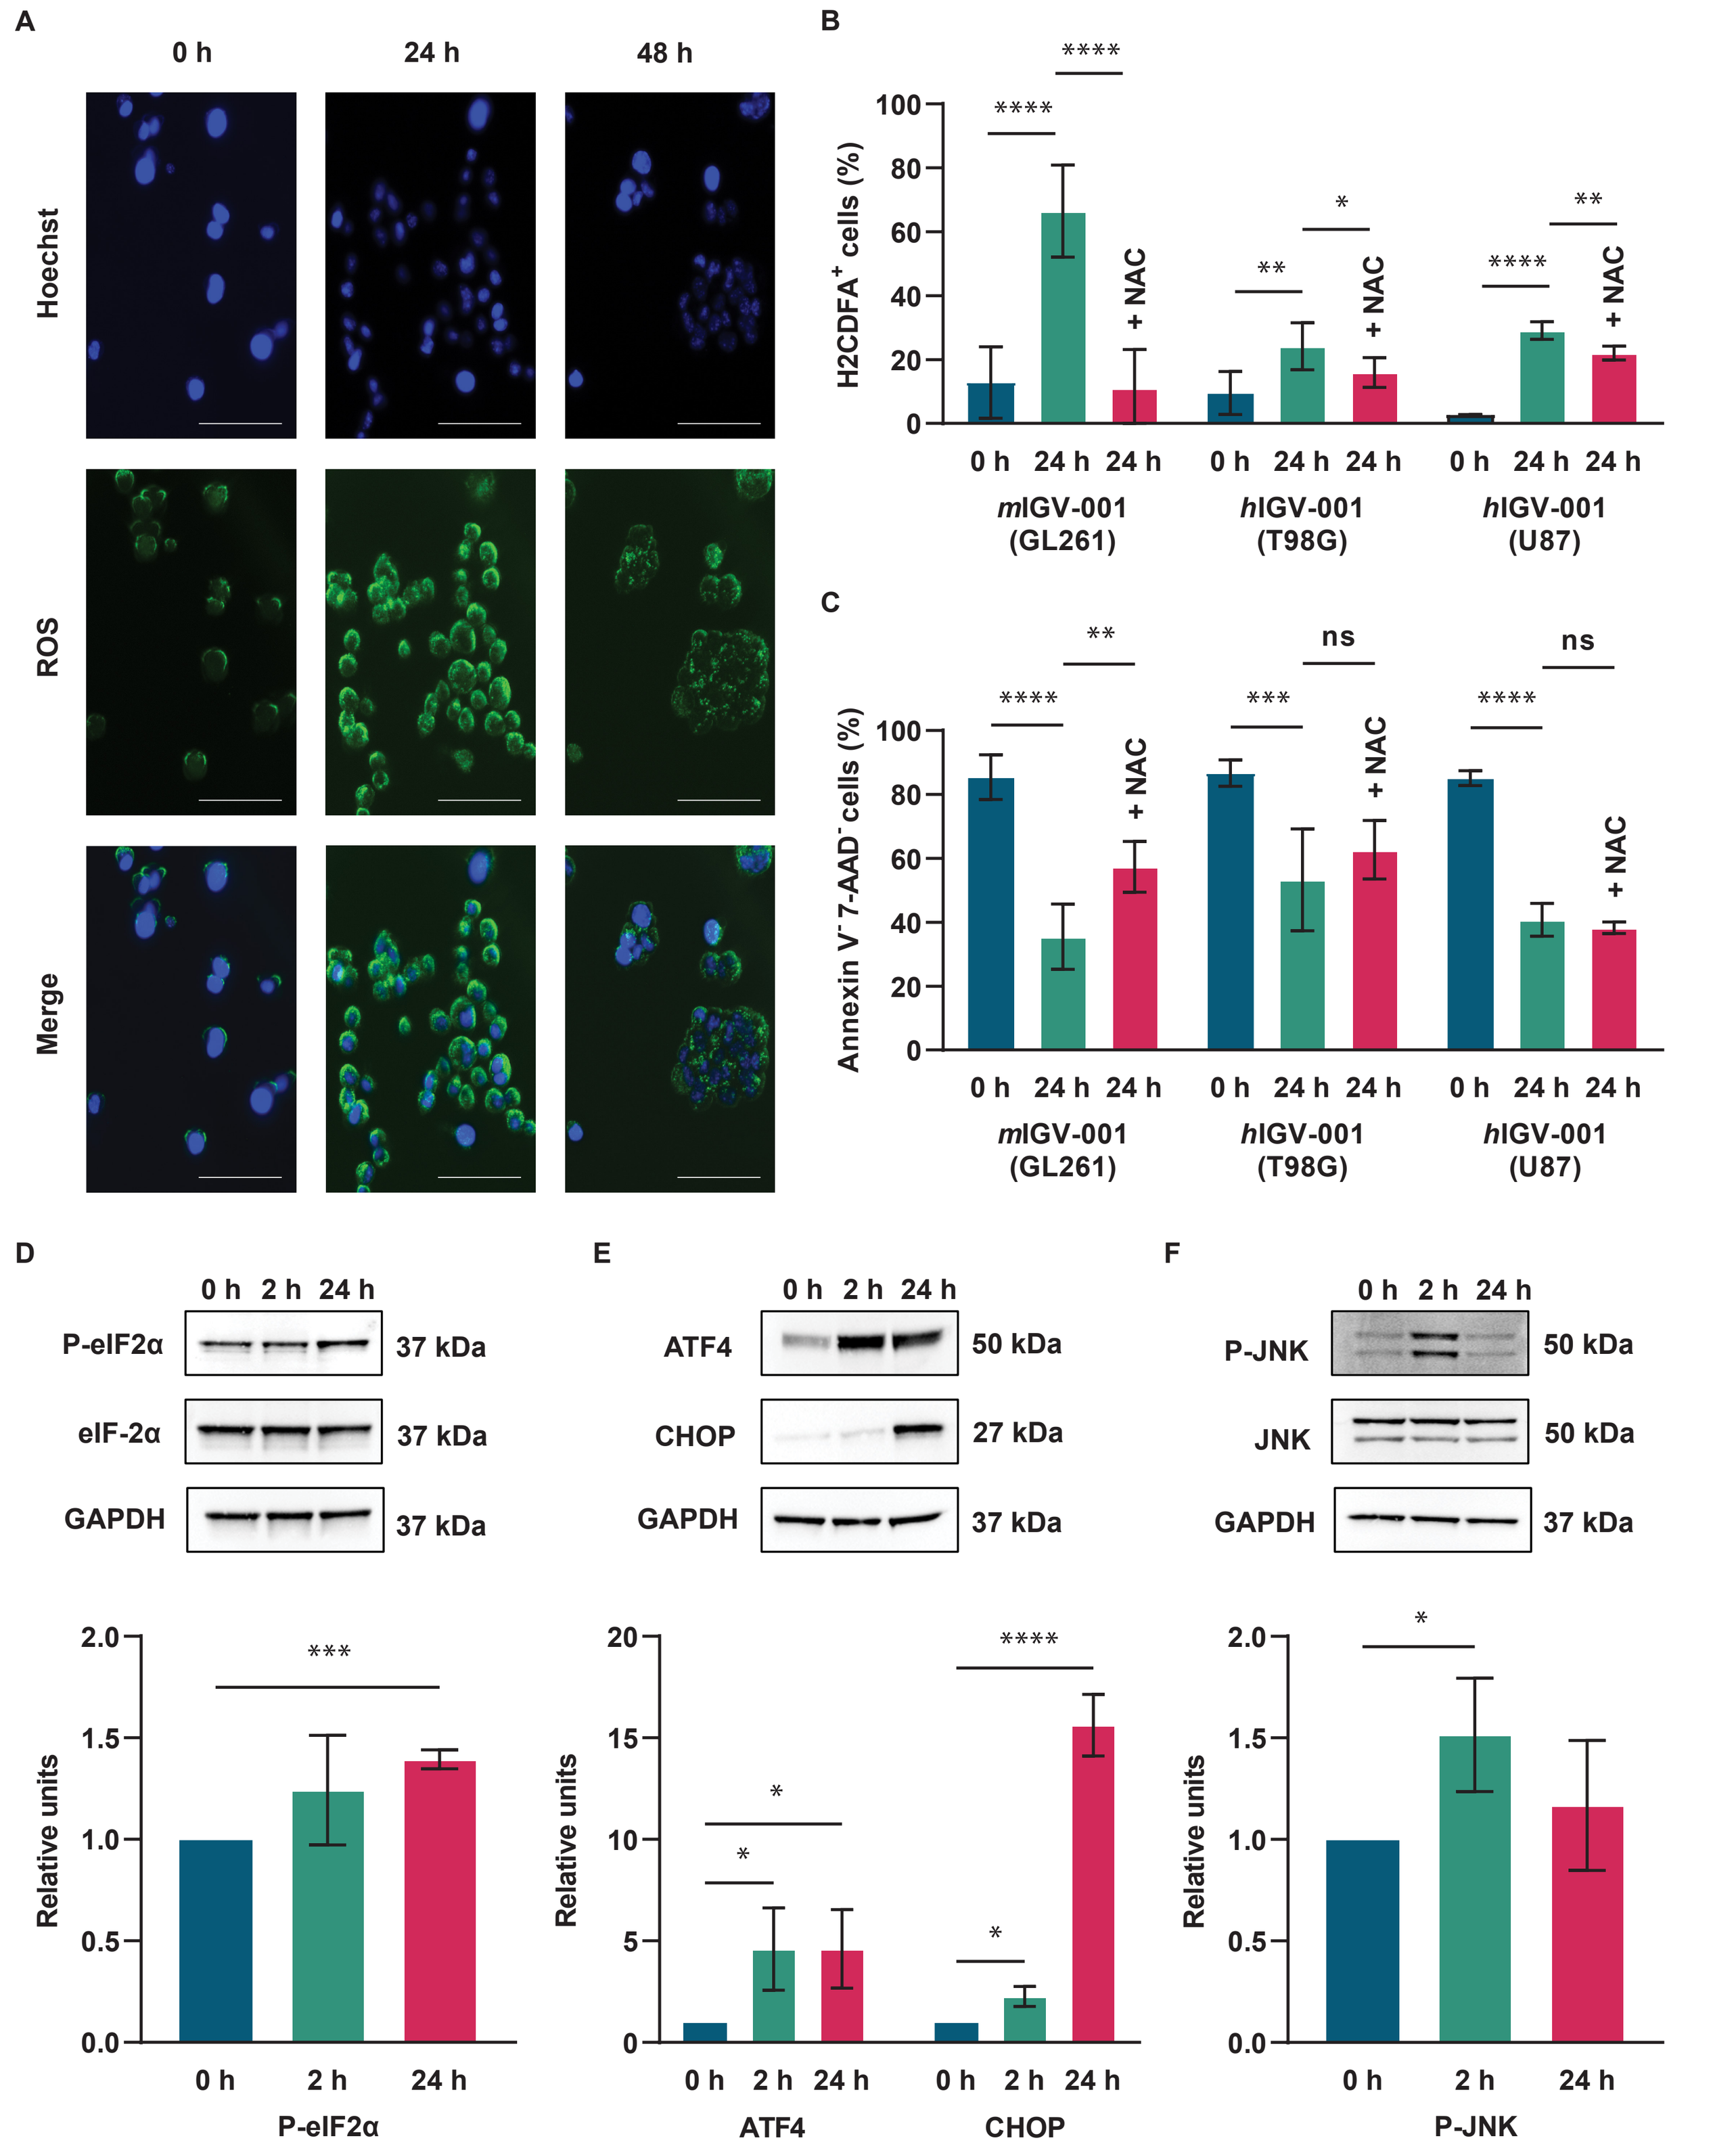
A
0 h
24 h
48 h
Hoechst
ROS
Merge
B
H2CDFA+ cells (%)
C
Annexin V-7-AAD- cells (%)
+ NAC
0 h 24 h 24 h
mIGV-001 (GL261)
hIGV-001 (T98G)
hIGV-001 (U87)
D
P-eIF2α 37 kDa
eIF-2α 37 kDa
GAPDH 37 kDa
E
ATF4 50 kDa
CHOP 27 kDa
GAPDH 37 kDa
F
P-JNK 50 kDa
JNK 50 kDa
GAPDH 37 kDa
0 h 2 h 24 h
Relative units
P-eIF2α
ATF4
CHOP
P-JNK

Figure 2. *m*IGV-001 was manufactured and cultured for 24 h. In order to quantify oxidative stress, cells were retrieved from the BDCs, stained with H2CDFA dye, and analyzed via flow cytometry or imaged using fluorescence microscopy. Where indicated, cultures were treated with NAC (1 or 2 mM). In separate experiments, retrieved cells were used for immunoblotting to detect ER stress markers. **(A)** Representative fluorescence microscopy images of ROS accumulation within *m*IGV-001 GBM cells. **(B)** Flow cytometric quantification of ROS accumulation in *m*- or *h*IGV-001 with **(C)** corresponding cell viability in the presence of the ROS scavenger, NAC. Western blot analyses of integrated stress response markers **(D)** eIF2a, **(E)** ATF4, and CHOP. **(F)** Activation of the ROS-sensitive effector kinase JNK. Western blot images are representative of 1 experiment out of 3 independent runs. Average normalized band densitometry for eIF2α, ATF4, and CHOP are plotted below the corresponding images. Results are presented as mean ± SD of 3 independent experiments with 2-3 BDCs per condition. Magnification = 40×. Scale bar = 75 mm. Statistical analysis was performed using either a standard unpaired or Welch's corrected t-test, *p<0.05, **p<0.01, ***p<001, and ****p<0.0001.

FIGURE 3

Cellular debris and particle diffusion in IGV-001

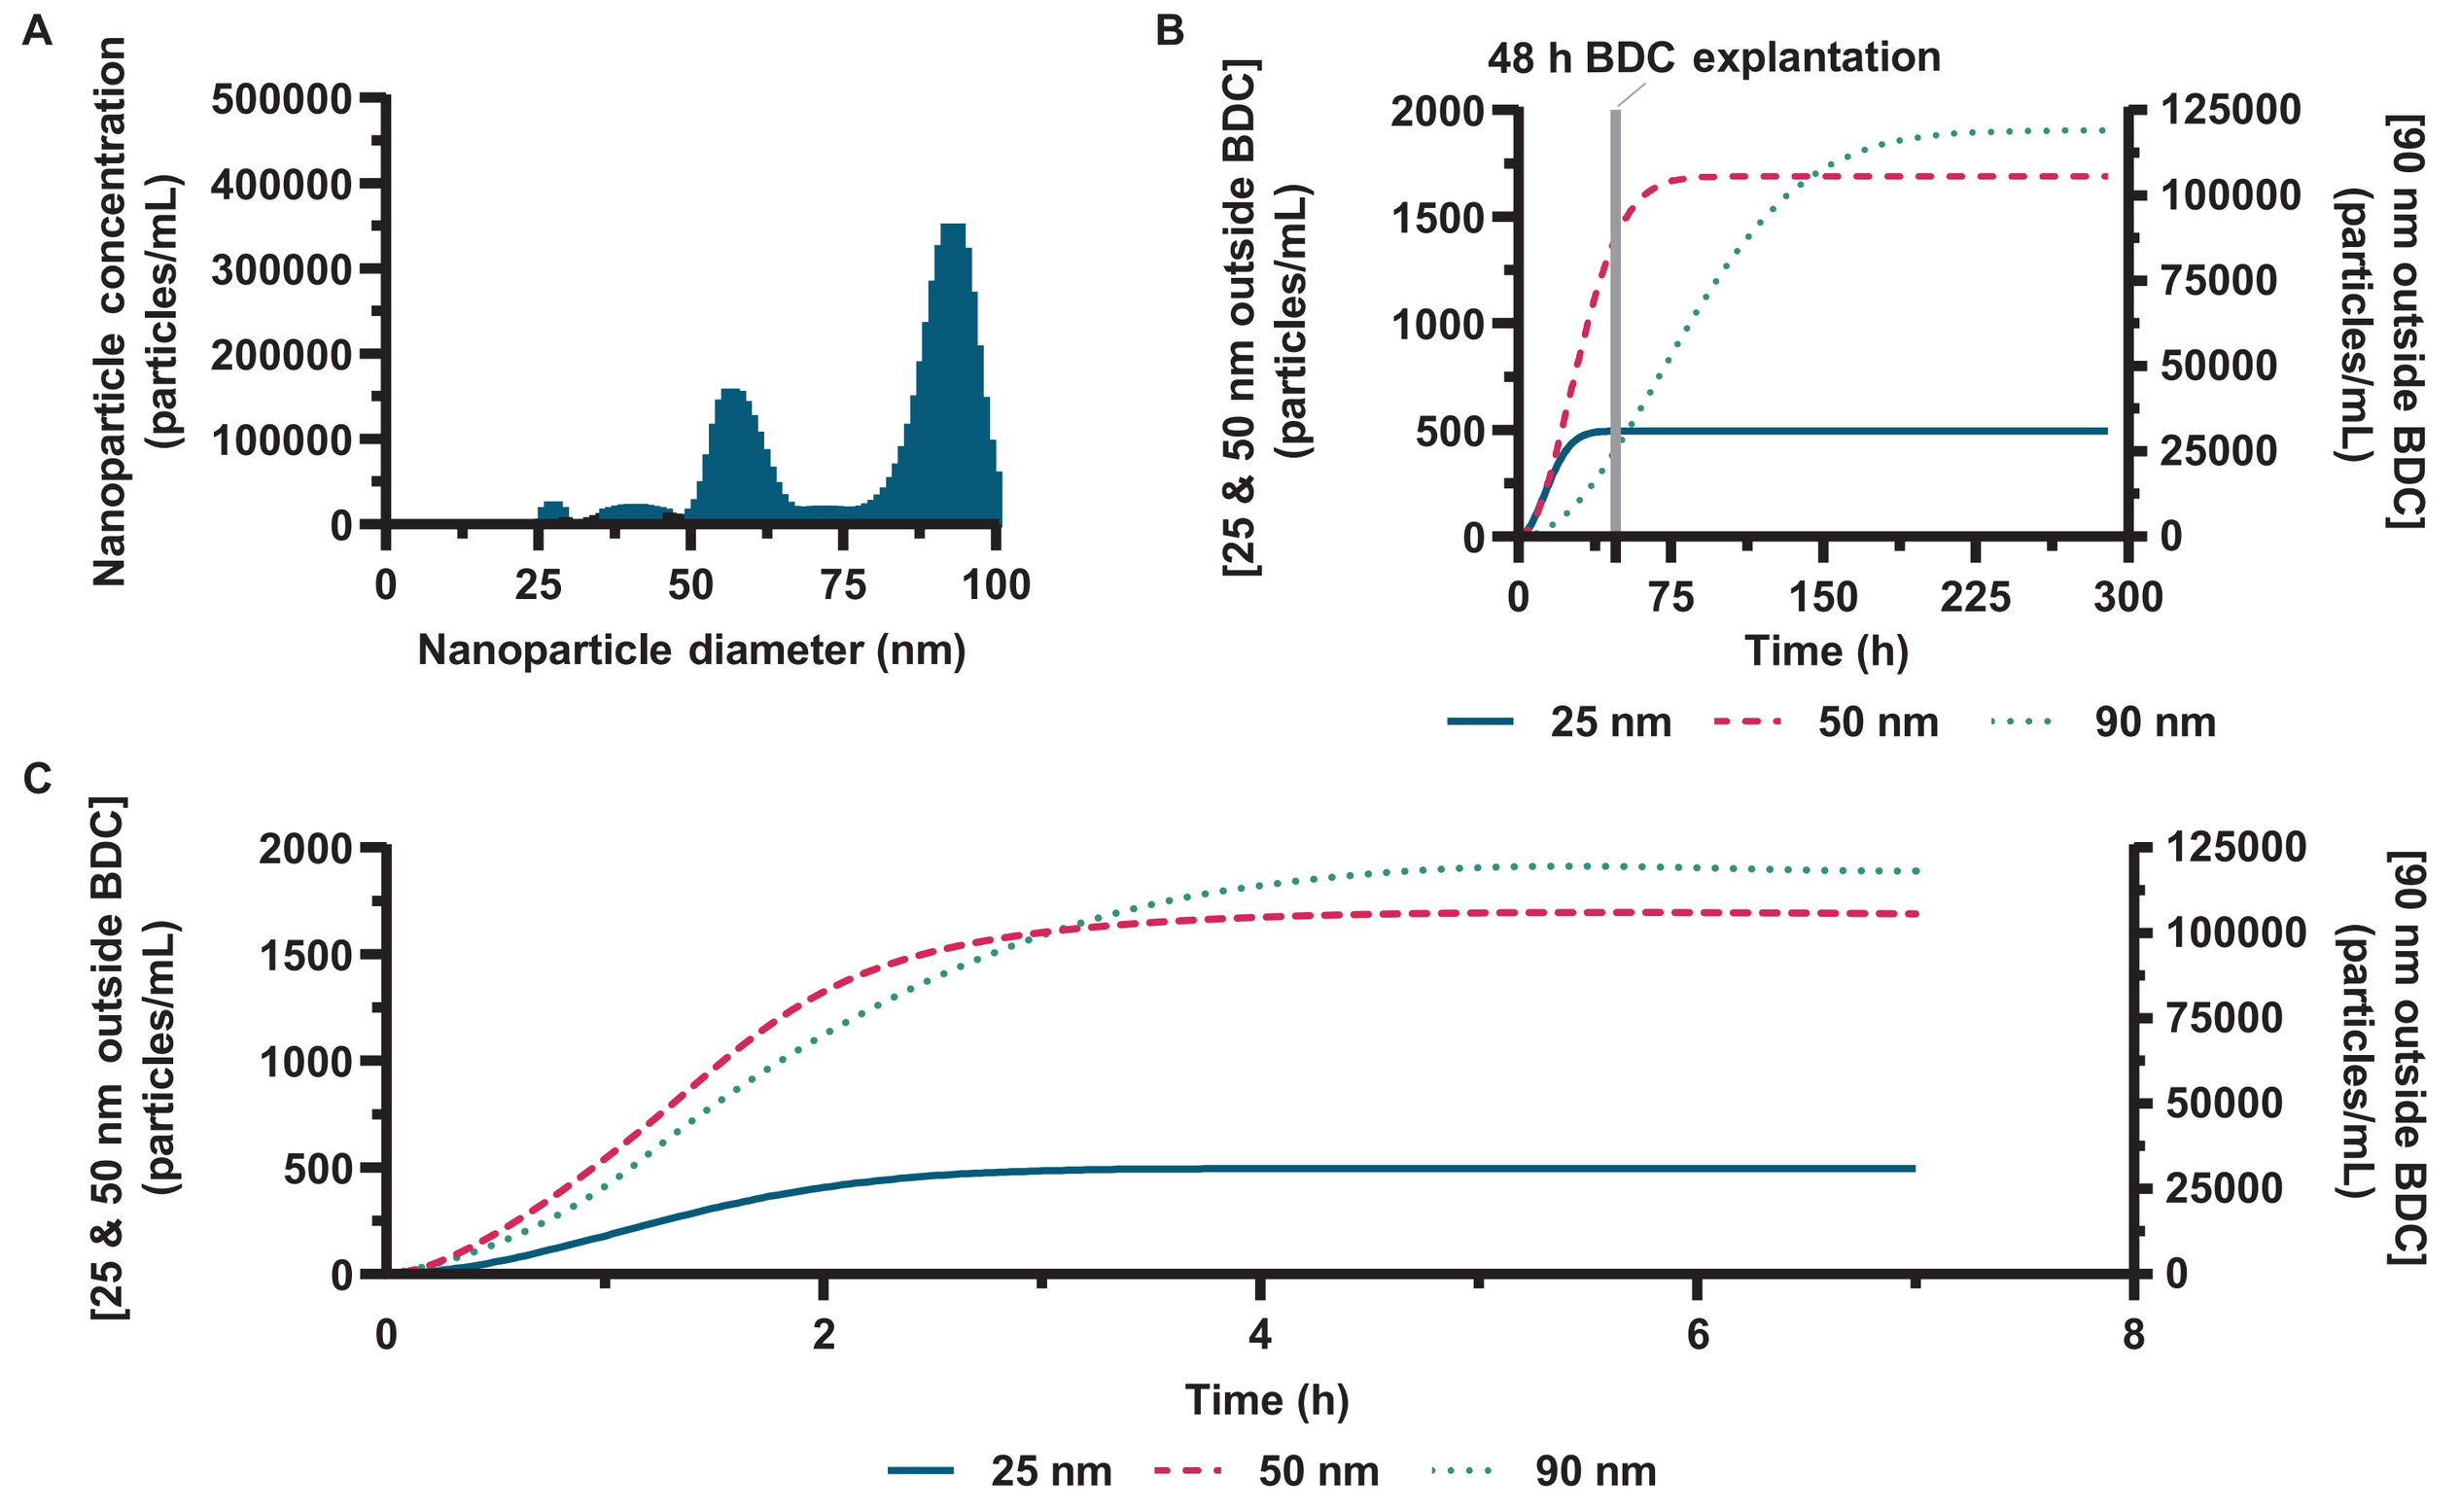
A
Nanoparticle concentration (particles/mL)
Nanoparticle diameter (nm)
B
48 h BDC explantation
[25 & 50 nm outside BDC] (particles/mL)
[90 nm outside BDC] (particles/mL)
Time (h)
25 nm
50 nm
90 nm
C

Figure 3. Particle concentration in the supernatant of *h*IGV-001 (T98G) BDC content after in vitro culture for 48 h was analyzed on a NanoSight NS300 (Malvern Panalytical) nanoparticle tracking analysis instrument. Nanoparticle transport was modeled using the Kedem–Katchalsky system of equations for solute transport kinetics across semipermeable membranes in MATLAB® 2022a (Mathworks™). **(A)** Distribution of tumor particle sizes in *h*IGV-001(T98G) BDCs supernatant from T98G GBM cells. **(B)** Theoretical concentrations of 25, 50, and 90 nm particle diffusion under passive conditions. **(C)** Theoretical concentrations of 25, 50, and 90 nm particle diffusion under dynamic conditions.

CONCLUSIONS

The use of IGV-001, a surgically implantable drug-device combination product, induces ROS-associated immunogenic cell death and the generation of a tumor-derived immunogenic payload that can be taken up by local dendritic cells, which travel to the proximal-draining lymph nodes to activate anti-tumor T cells. These results support the mechanism of action of IGV-001, which is currently being studied in a randomized Phase 2b trial in newly diagnosed glioblastoma [NCT04485949].

DISCLOSURES

CC, KK, EE, CU, AZ, ME, and JZ are current or former employees of IMVAX, Inc.

REFERENCES

1. Andrews, DW, et al. *Clin Cancer Res.* 2021;27(7):1912-1922.
2. Stupp, R, et al. *N Engl J Med.* 2005;352(10):987-96.
3. Zilberberg, J, et al. *J Immunother Cancer.* 2021;9(Suppl 2):A231.
4. Cultrara, C, et al. *J Immunother Cancer.* 2022;10(Suppl 2):A1–A1595.

Poster presented at the American Association of Cancer Research Annual Meeting 2023. April 14-19, 2023, Orlando, FL USA.

© Copyright 2023 IMVAX®, Inc. All rights reserved.