Personalized Immunotherapeutic Platform, with Evidence of Clinical Activity in Glioblastoma, Protects Mice Against Ovarian, Liver & Bladder Cancer Tumor Challenges

Amelia Zellander[1], Christopher Uhl[1], Christopher Cultrara[1], Kenneth Kirby[1], Essam Elrazaq[1], Charles B. Scott[2,3], David W. Andrews[1,3], Jenny Zilberberg[1], Mark A. Exley[1]

[1]Imvax Inc., [2]CBS Squared Inc., [3]Thomas Jefferson University, Philadelphia, PA.

#1398

BACKGROUND

Imvax, Inc. is developing a novel personalized immunotherapeutic platform combining irradiated patient-derived tumor cells and insulin-like growth factor type-1 receptor antisense oligonucleotide (IMV-001) in biodiffusion chambers (BDCs; 0.1-micron pores). The glioblastoma (GBM) drug-device combination product, IGV-001, was evaluated in a newly diagnosed GBM phase 1b clinical trial[1]. Median overall survival of highest exposure IGV-001-treated 'Stupp-eligible'[2] patients (n=10) was 38.2 months compared with 16.2 months in recent standard-of-care-treated patients (P=0.044)[1] [NCT02507583]. Imvax also reported anti-tumor activity of mIGV-001 (murine equivalent IGV-001) in the GL261-luc GBM murine model[3]. Here we show activity with the equivalent murine approach in multiple cancer models, highlighting the transformative potential of this immunotherapeutic platform beyond glioblastoma.

METHODOLOGY

We utilized ID8-luciferase (-Luc) intraperitoneal ovarian cancer, Hepa1-6-Luc orthotopic hepatocellular carcinoma, and MBT-2 orthotopic urothelial murine cancer models. One BDC containing saline or 1×10^6 IMV-001-treated tumor cells (hereinafter, *m*IOC-001, *m*IHC-001, and *m*IUC-001, respectively) was implanted in flanks of mice and explanted 48 h later, as per glioblastoma clinical protocol. Primary tumor challenge was conducted 28 d after chamber implantation. Intra-mammary rechallenge in the Hepa1-6 model was conducted 102 d after orthotopic challenge alongside naive controls, since no primary controls were still alive. Mice were monitored for survival and tumor growth. MSD (meso scale discovery) cytokine analysis was conducted on serum. Splenocytes and PBMCs were analyzed with flow cytometry.

FIGURE 1

Murine model methods[4-7]

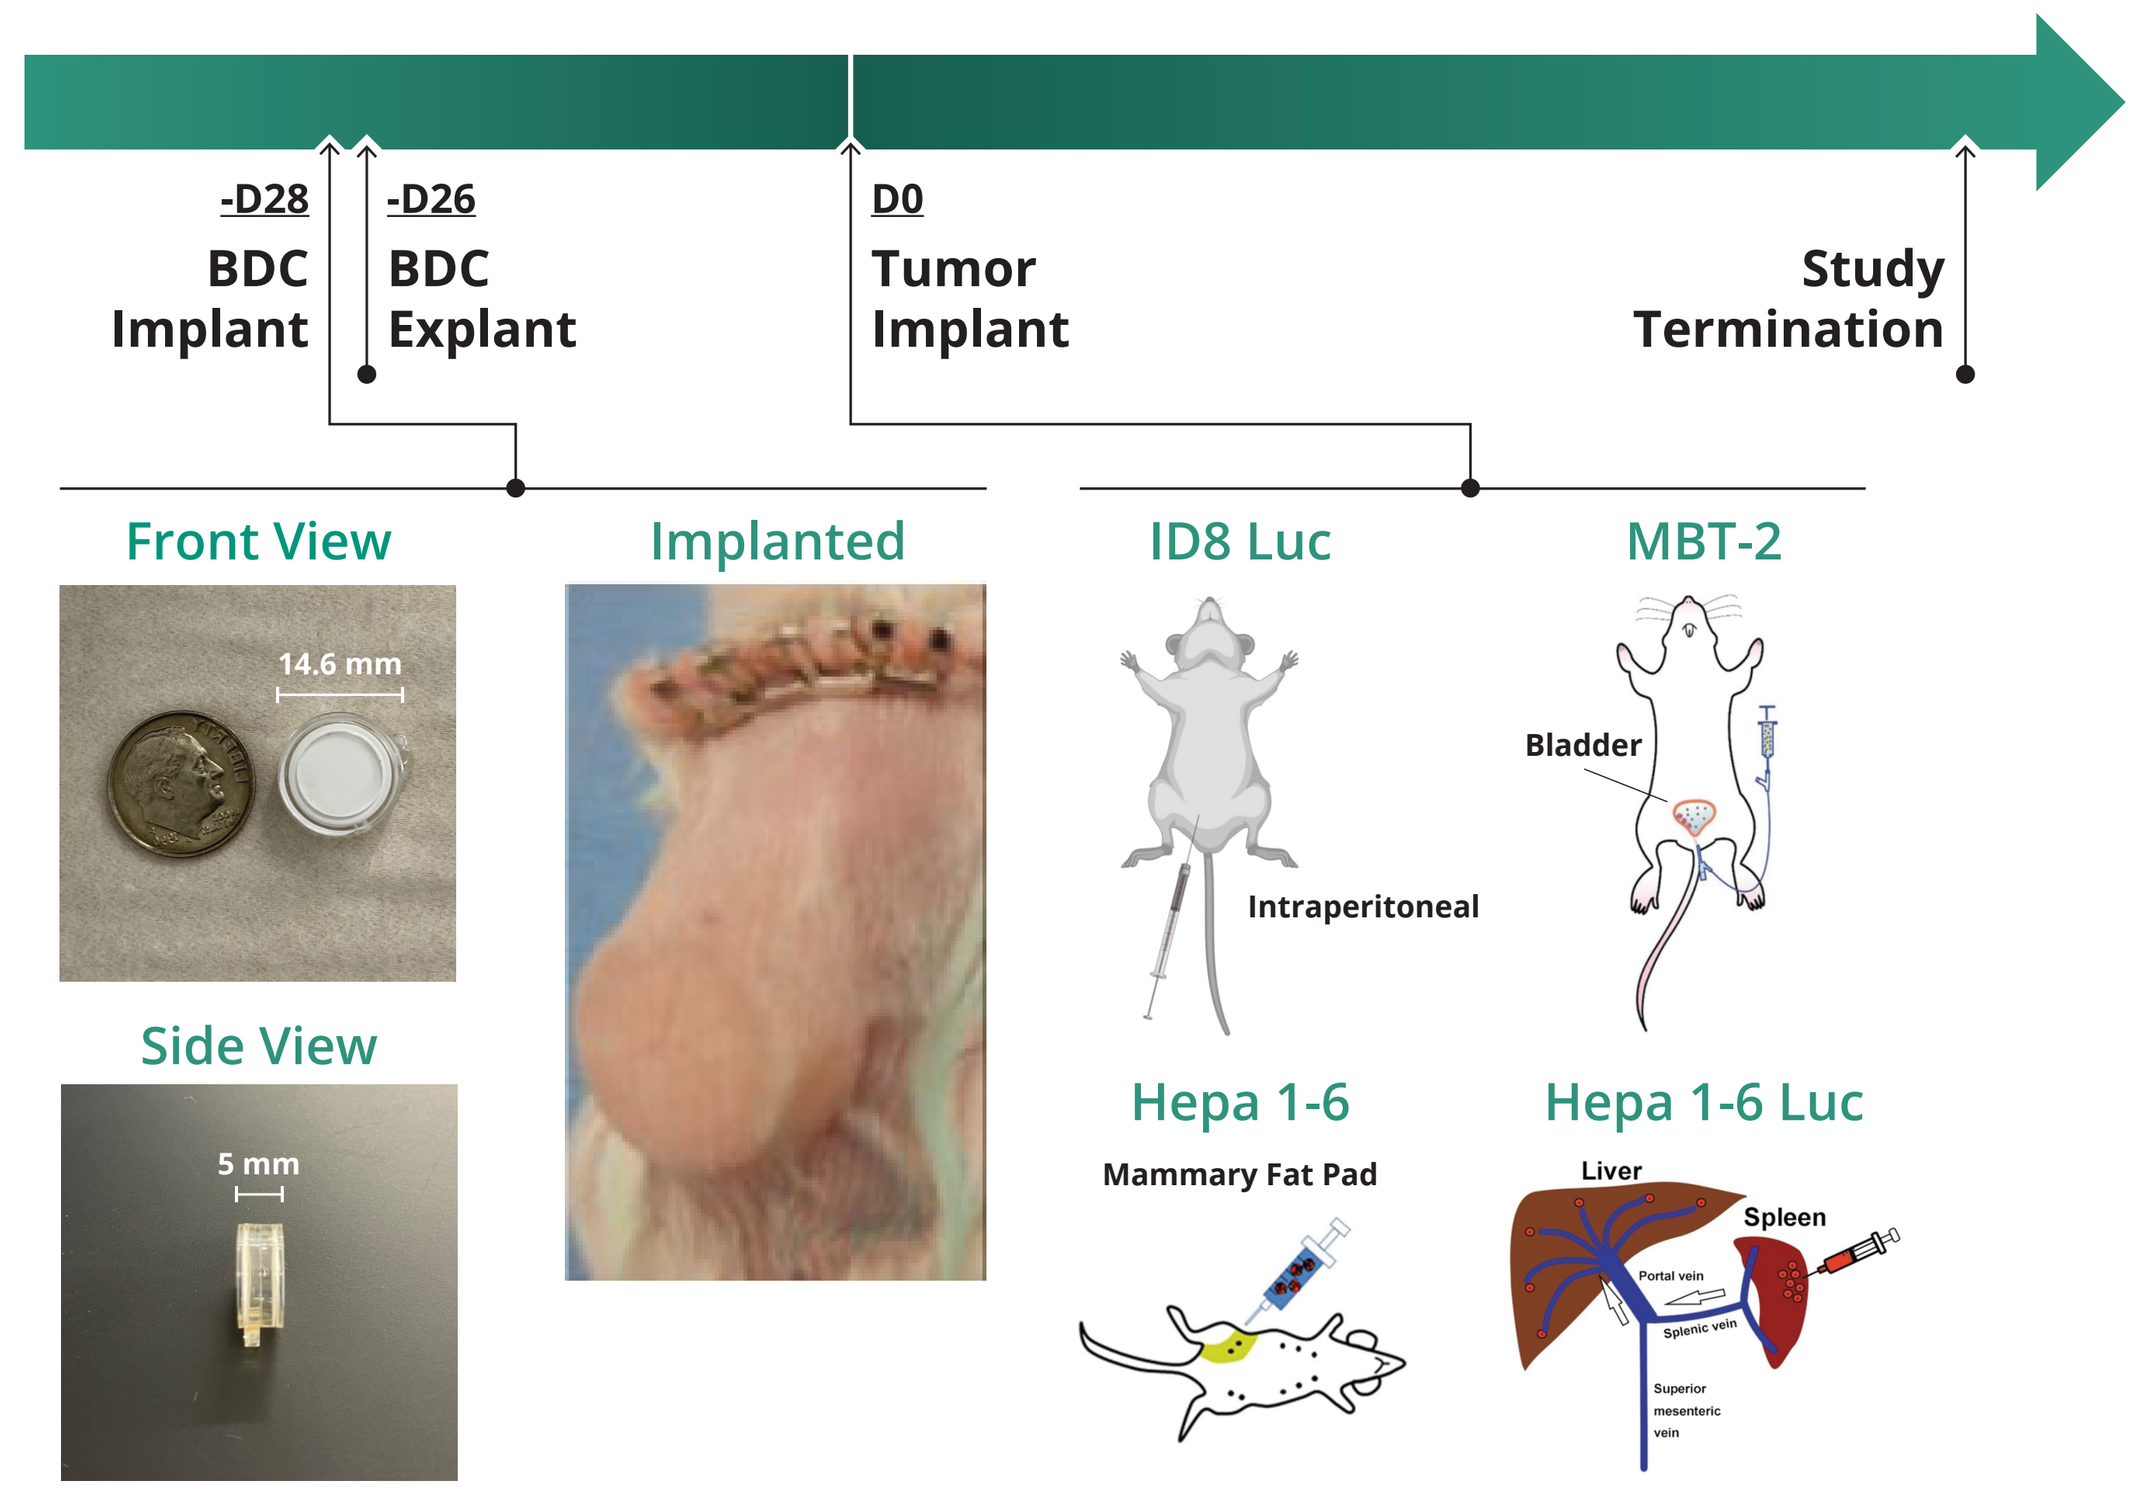
-D28
BDC Implant
-D26
BDC Explant
D0
Tumor Implant
Study Termination
Front View
14.6 mm
Implanted
Side View
5 mm
ID8 Luc
Intraperitoneal
MBT-2
Bladder
Hepa 1-6
Mammary Fat Pad
Hepa 1-6 Luc
Liver
Spleen

RESULTS

- Improved post-treatment survival and reduced tumor size:
 - **Ovarian:** 60% of IOC-001-treated mice survived to end of study at D58 post–tumor challenge, compared to only 19% of mice in the saline control group (Median Survival Time [MST]=D37, P=0.0041). Treatment notably reduced tumor size as compared to the control (fig. 2).
 - **Liver:** 50% of IHC-001-treated mice survived beyond D110 post-tumor (MST=D60.5). There were no survivors in the primary control group beyond D28 (MST=D18; P=0.0041). Treatment notably reduced tumor size as compared to the control. Hepa1-6 intramammary rechallenge at D102 demonstrated durable systemic immunity in survivors (figs. 3 and 4).
 - **Bladder:** 42% of IUC-001-treated mice survived up to D90 post-tumor challenge (MST=D33); all control mice expired by D37 (MST=D21; P=0.0163). Surviving *m*IUC-001 treated mice had low end-of-life bladder weights (fig. 5).
- Circulating IFNγ was significantly higher in *m*IOC-001-treated mice compared to controls at D1 post-tumor challenge (fig. 2).
- Reduced PD1 expression in T cells of *m*IHC-001 treated mice.

FIGURE 2

Ovarian Cancer Model (ID8-Luc) - *m*IOC-001 Treatment

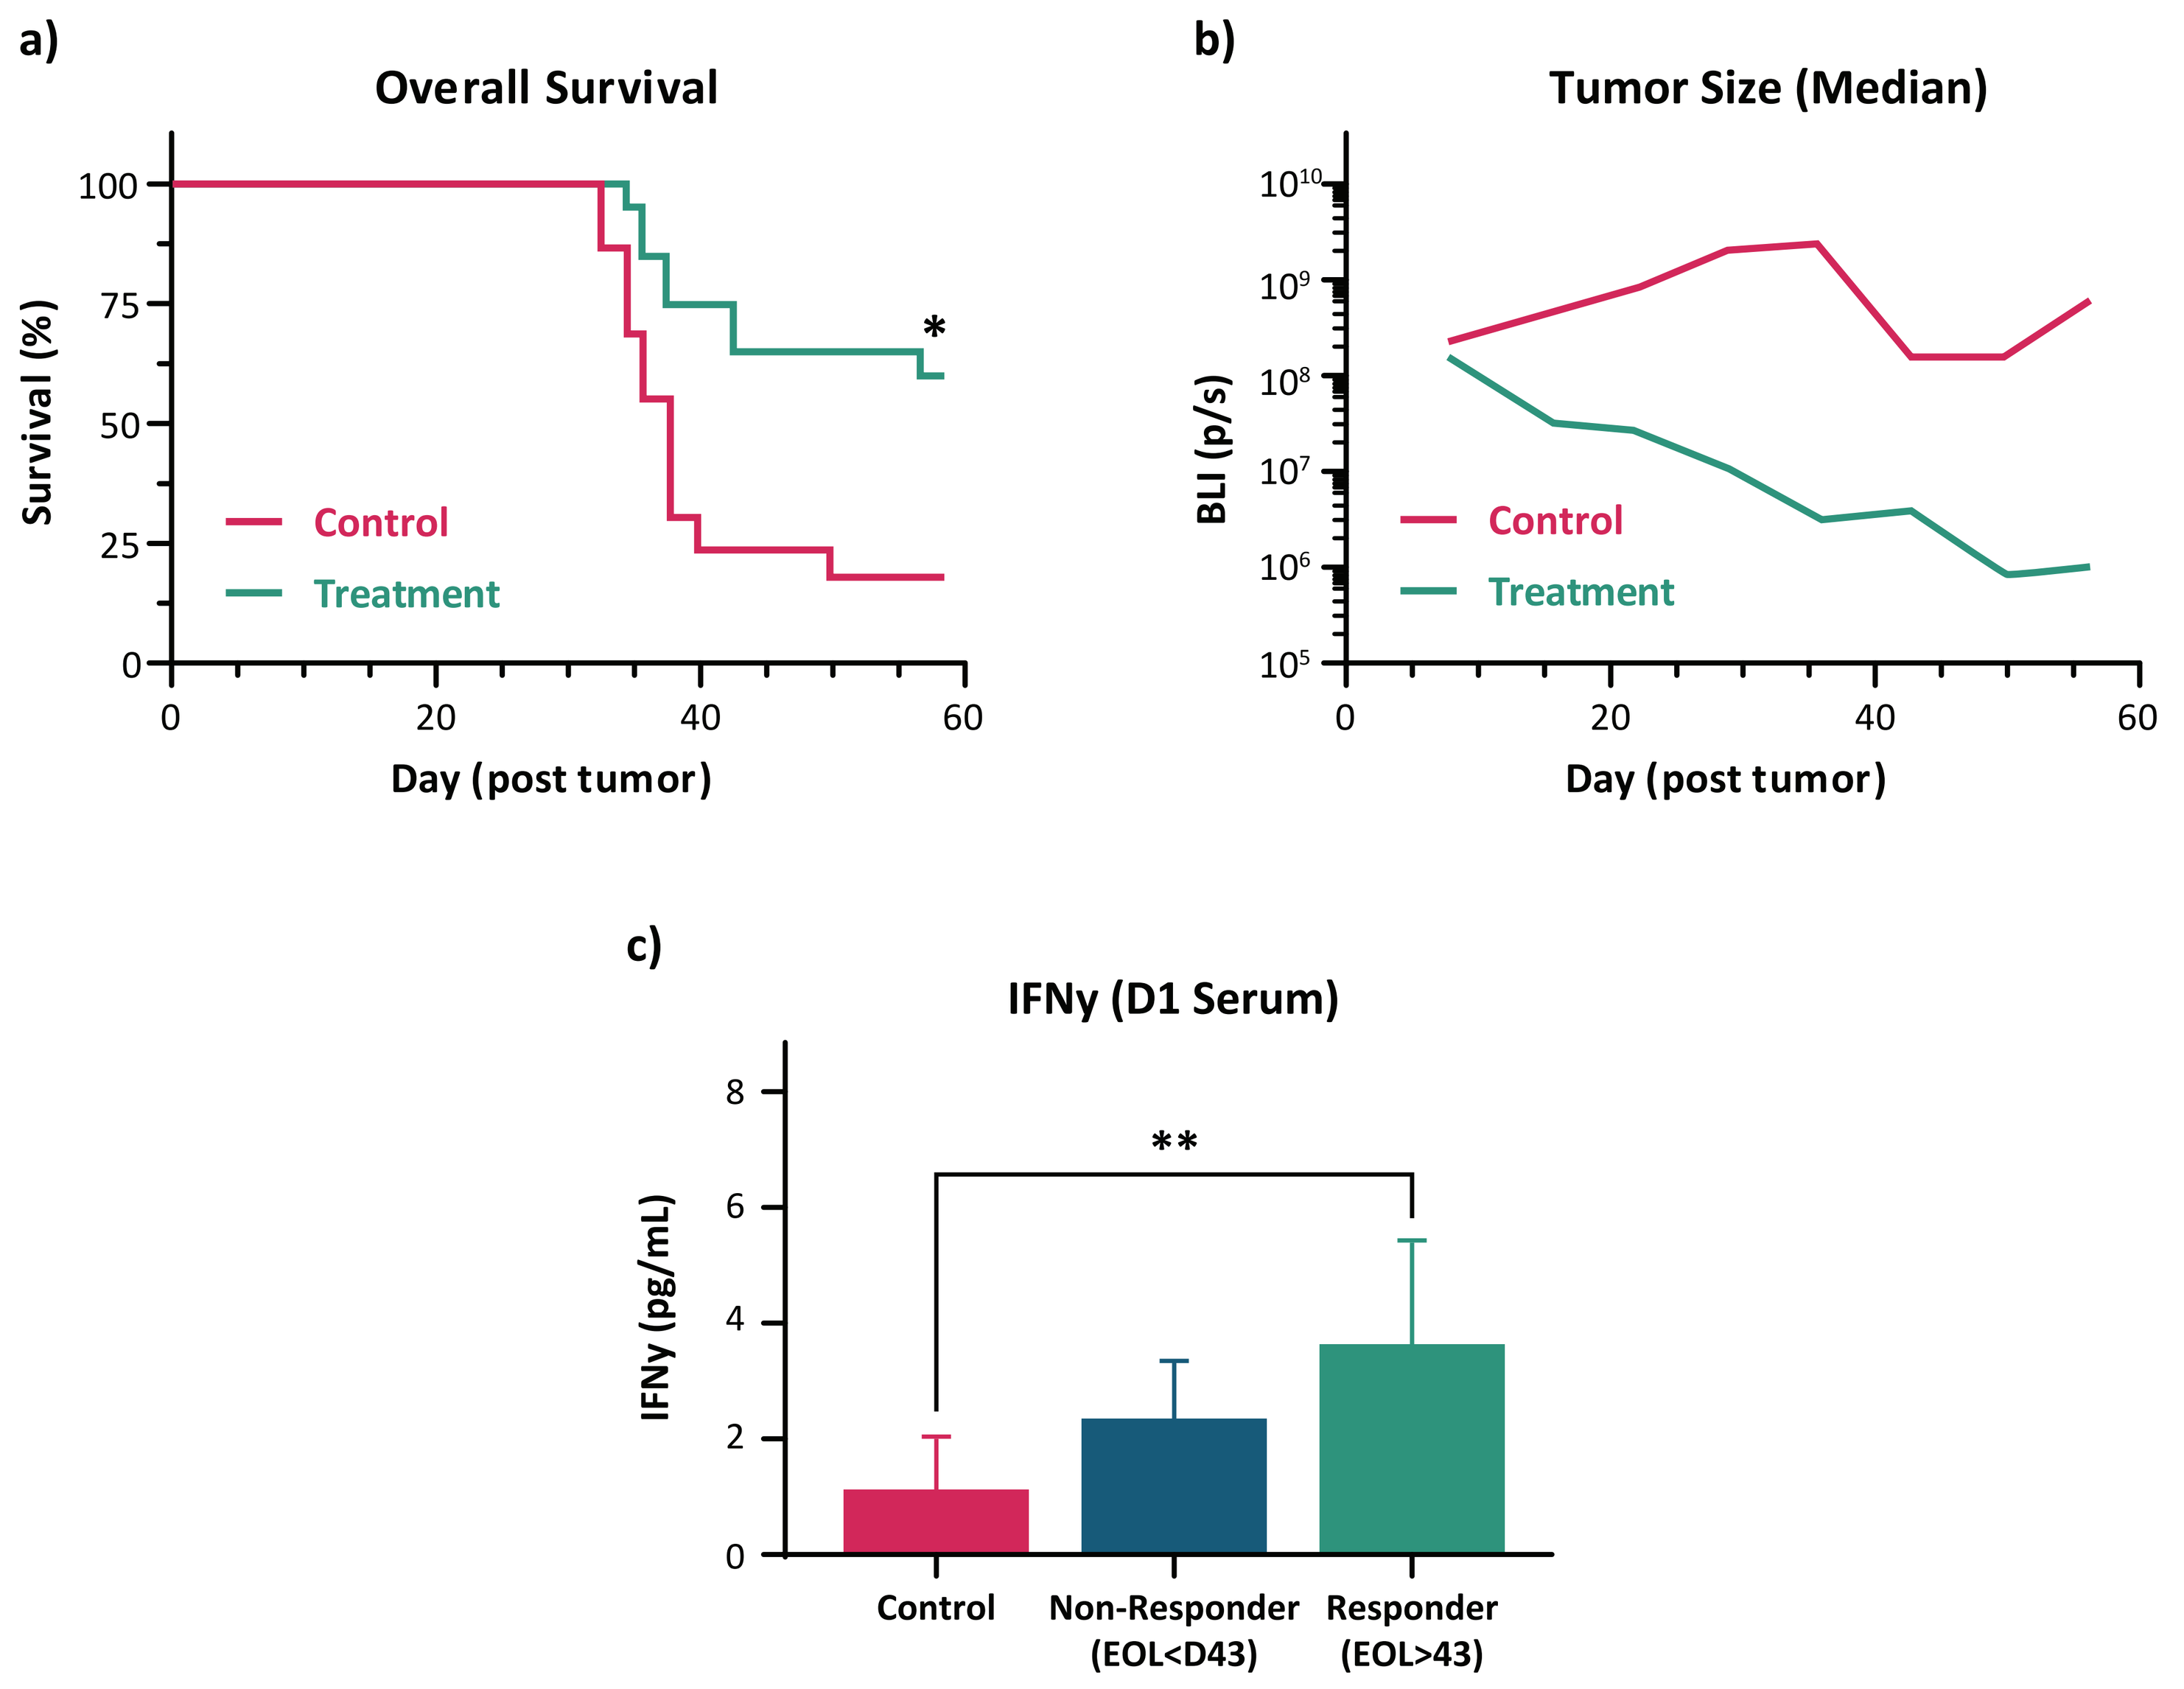
a)
Overall Survival
Survival (%)
Day (post tumor)
Control
Treatment
b)
Tumor Size (Median)
BLI (p/s)
Day (post tumor)
Control
Treatment
c)
IFNy (D1 Serum)
IFNy (pg/mL)
Control
Non-Responder (EOL<D43)
Responder (EOL>43)

Immunotherapy significantly improves **a)** survival and reduces **b)** tumor size in the murine ovarian cancer model (Control n=16; Treatment n=20). **c)** Increased IFNγ in immunotherapy treated mice suggests a greater Th1 response (Control n=16; Treated Non-Responder n=7; Treated Responder n=13). * P=0.0041, ** P<0.0001.

FIGURE 3

Liver Cancer Model (Hepa 1-6) - *m*IHC-001 Treatment

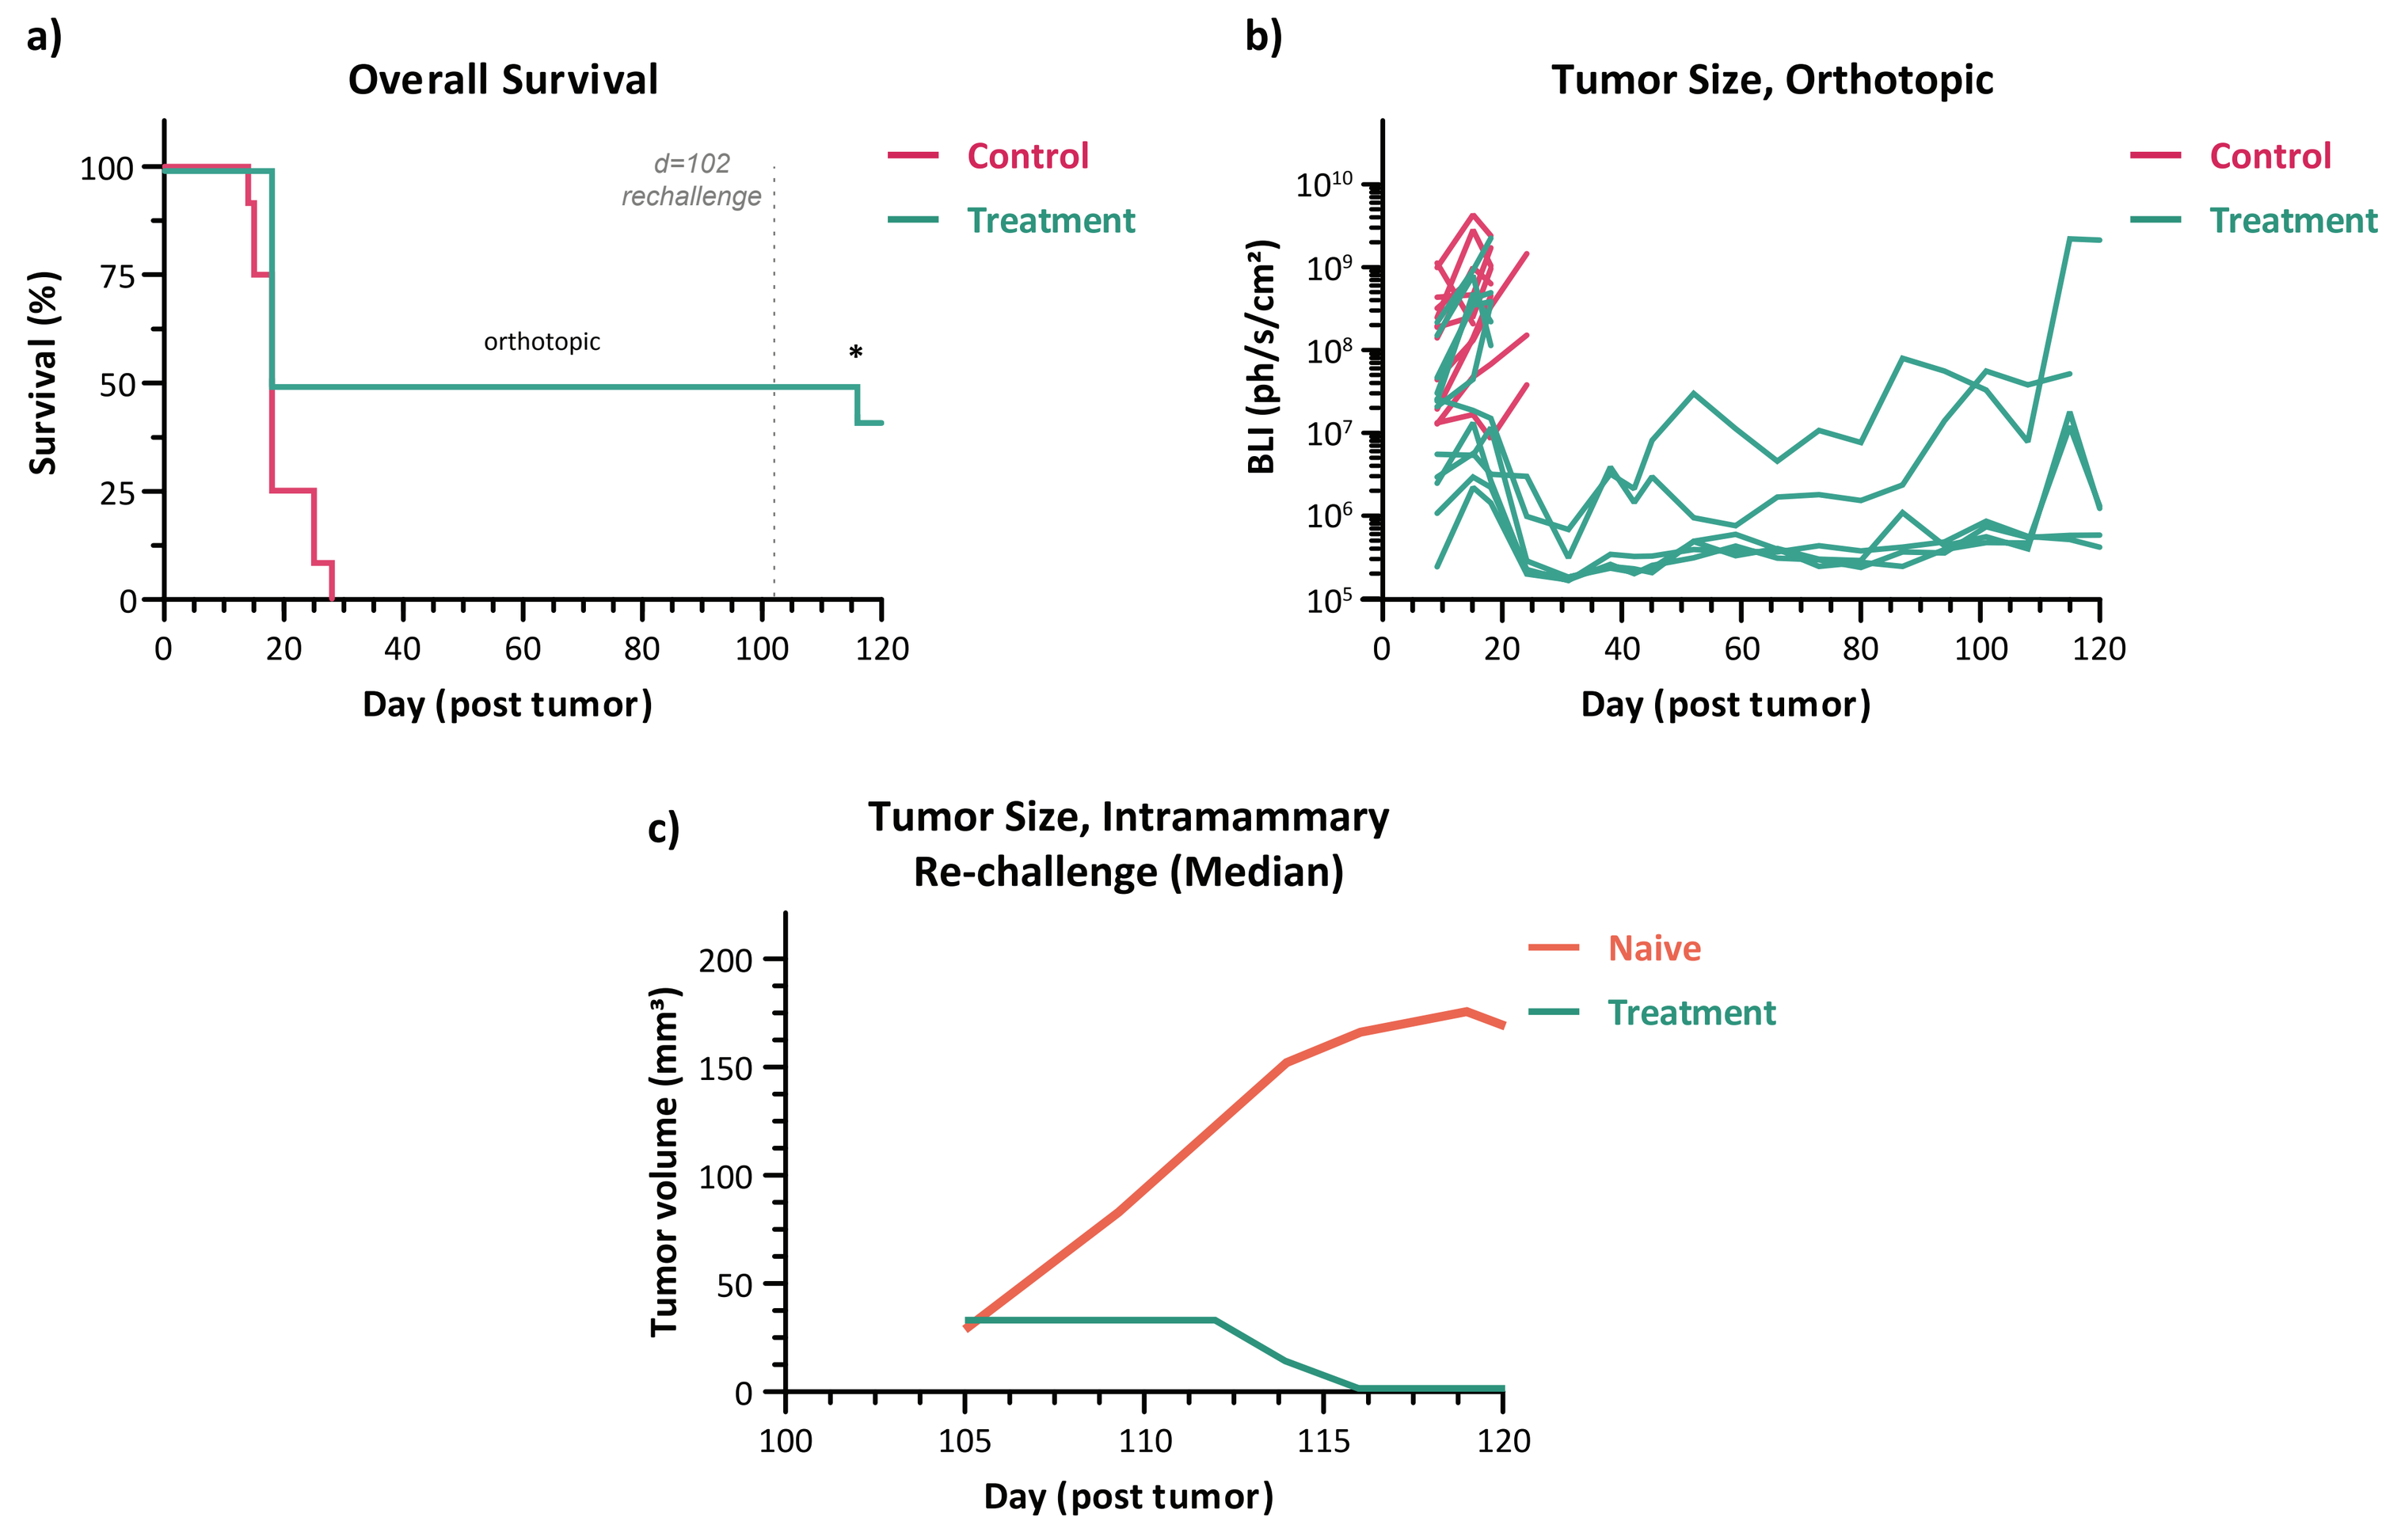
a)
Overall Survival
Survival (%)
d=102 rechallenge
orthotopic
Day (post tumor)
Control
Treatment
b)
Tumor Size, Orthotopic
BLI (ph/s/cm²)
Day (post tumor)
Control
Treatment
c)
Tumor Size, Intramammary Re-challenge (Median)
Tumor volume (mm³)
Day (post tumor)
Naive
Treatment

Immunotherapy significantly improves **a)** survival and reduces **b)**, **c)** tumor size in the murine liver cancer model (Control n=12; Treatment n=12; Naive n=6). * P=0.0041.

FIGURE 4

Liver Cancer Model (Hepa 1-6) - *m*IHC-001 Treatment

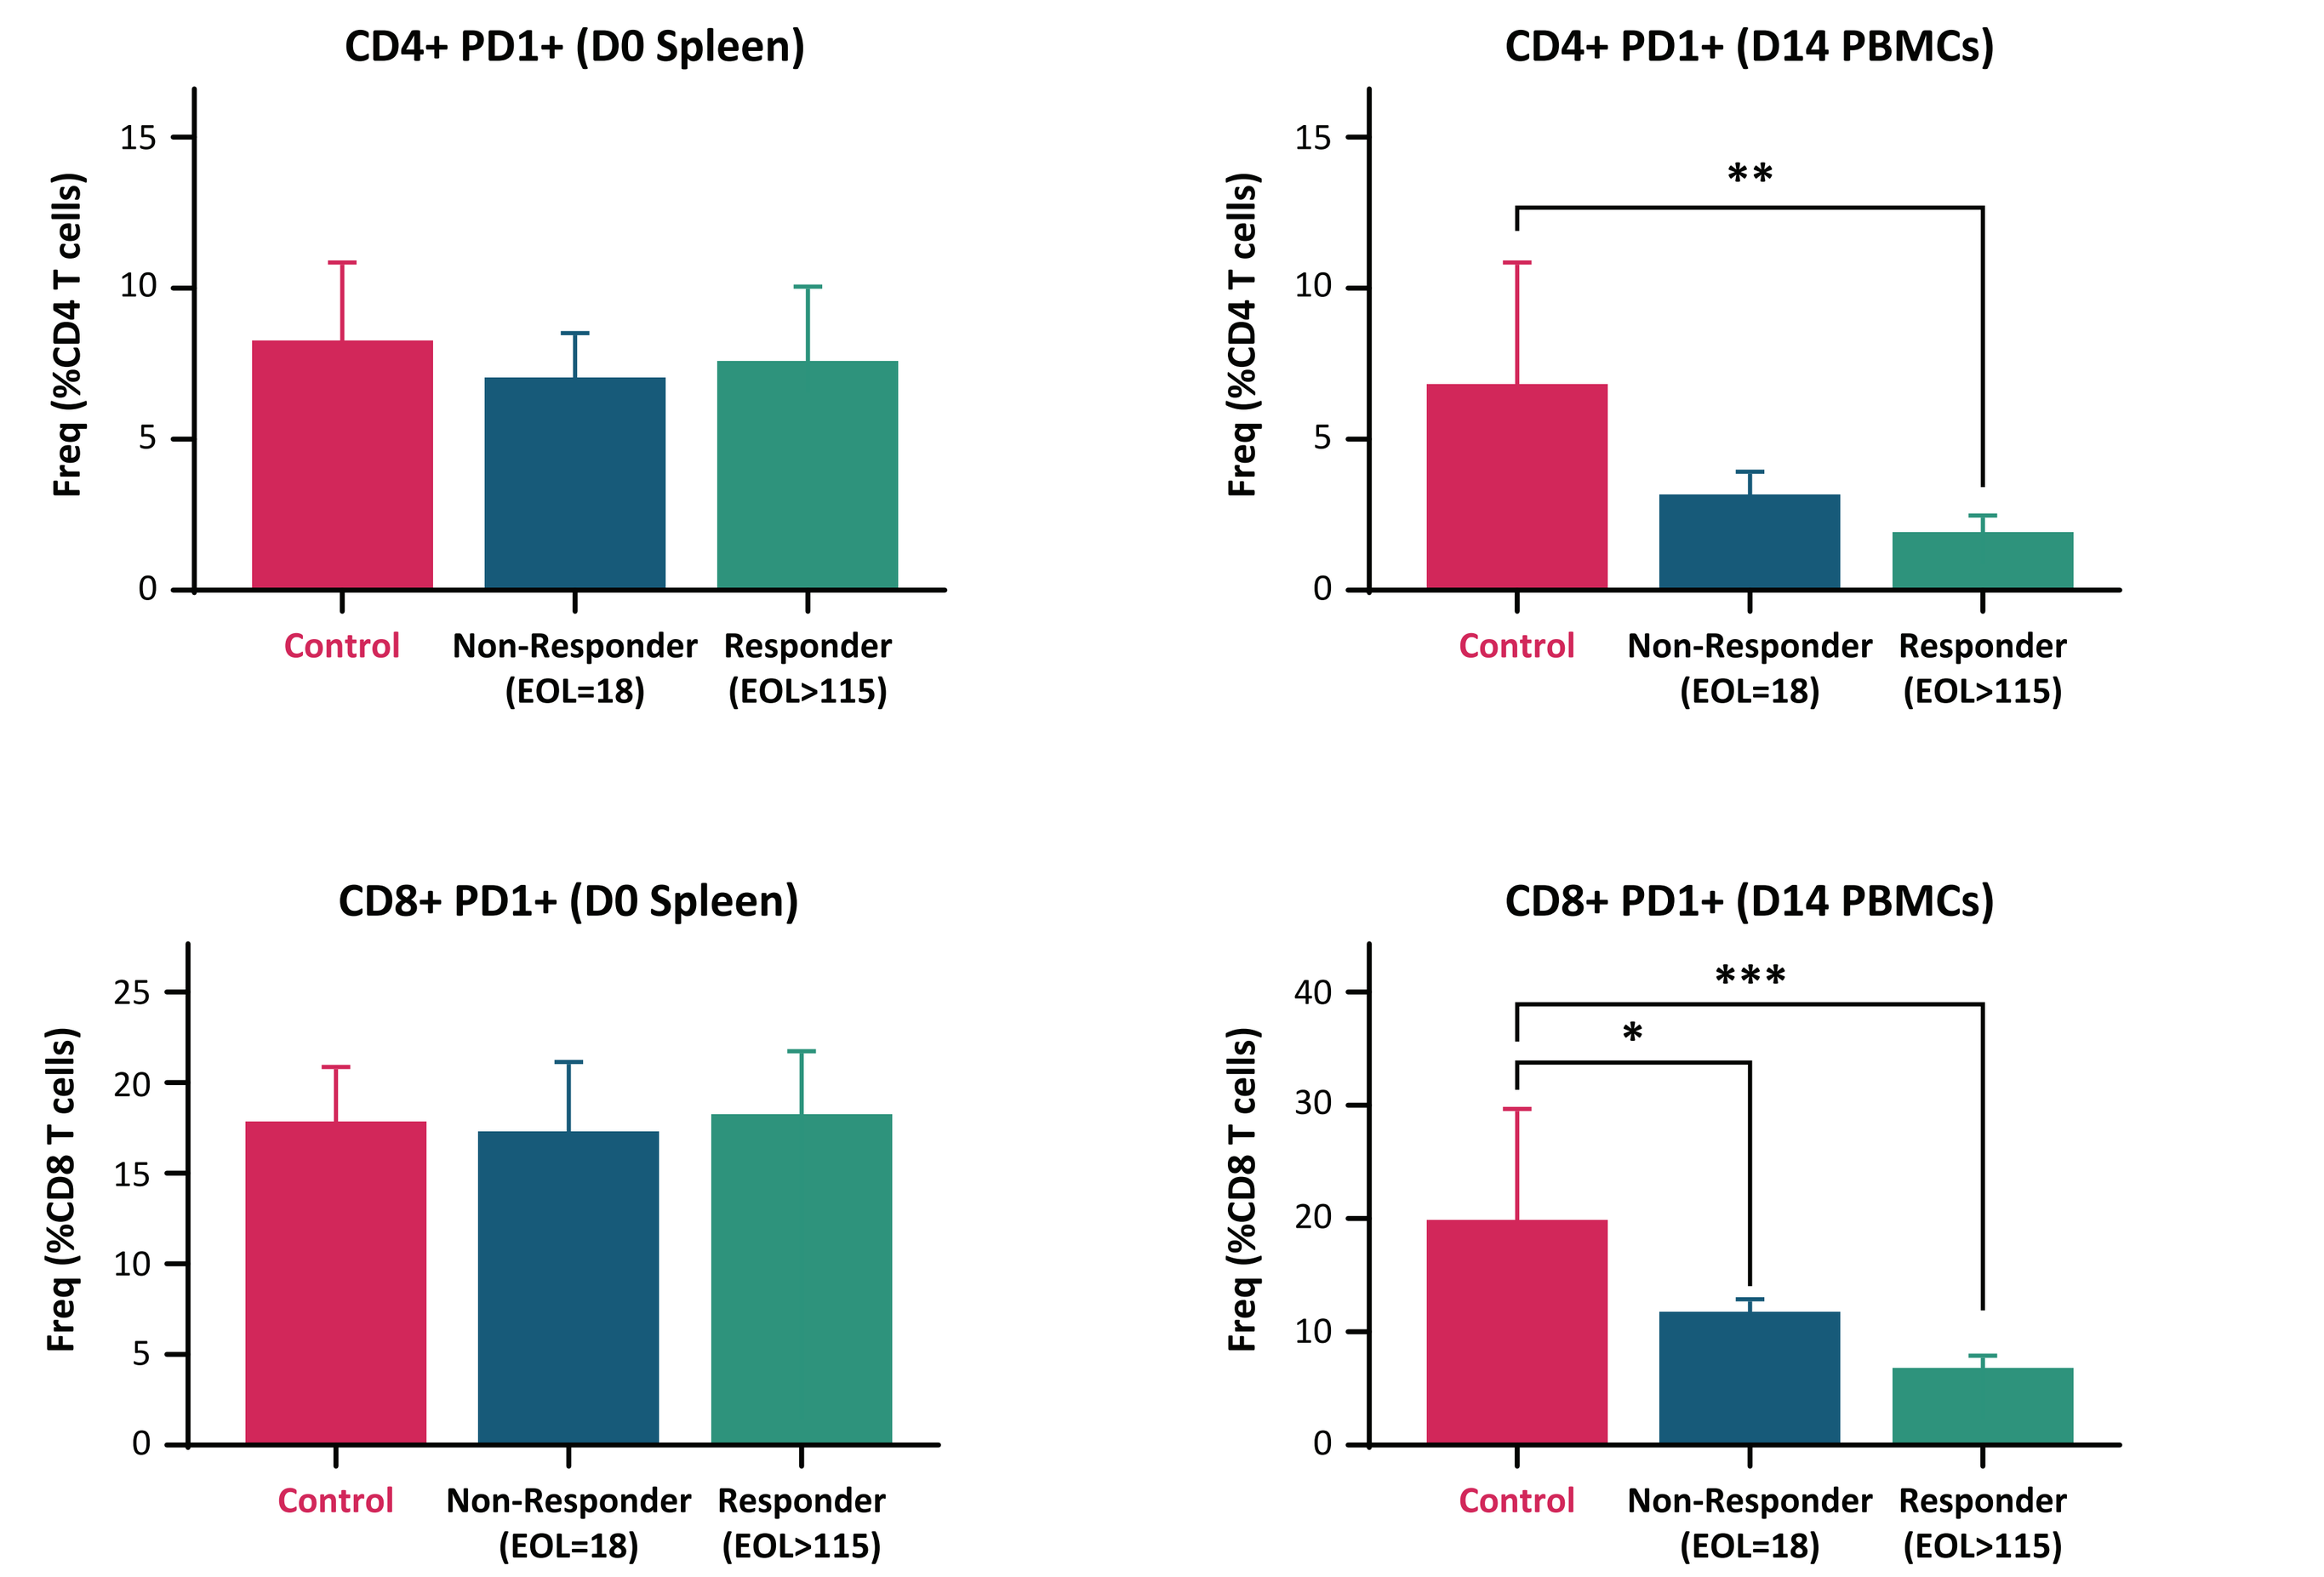
CD4+ PD1+ (D0 Spleen)
CD4+ PD1+ (D14 PBMCs)
Freq (%CD4 T cells)
CD8+ PD1+ (D0 Spleen)
CD8+ PD1+ (D14 PBMCs)
Freq (%CD8 T cells)
Control
Non-Responder (EOL=18)
Responder (EOL>115)

Post-treatment T cell activity in murine liver cancer model (Control n=12; Treated Non-Responder n=6; Treated Responder n=6). * P=0.0469, ** P=0.0162, *** P=0.0031.

FIGURE 5

Bladder Cancer Model (MBT-2) - *m*IUC-001

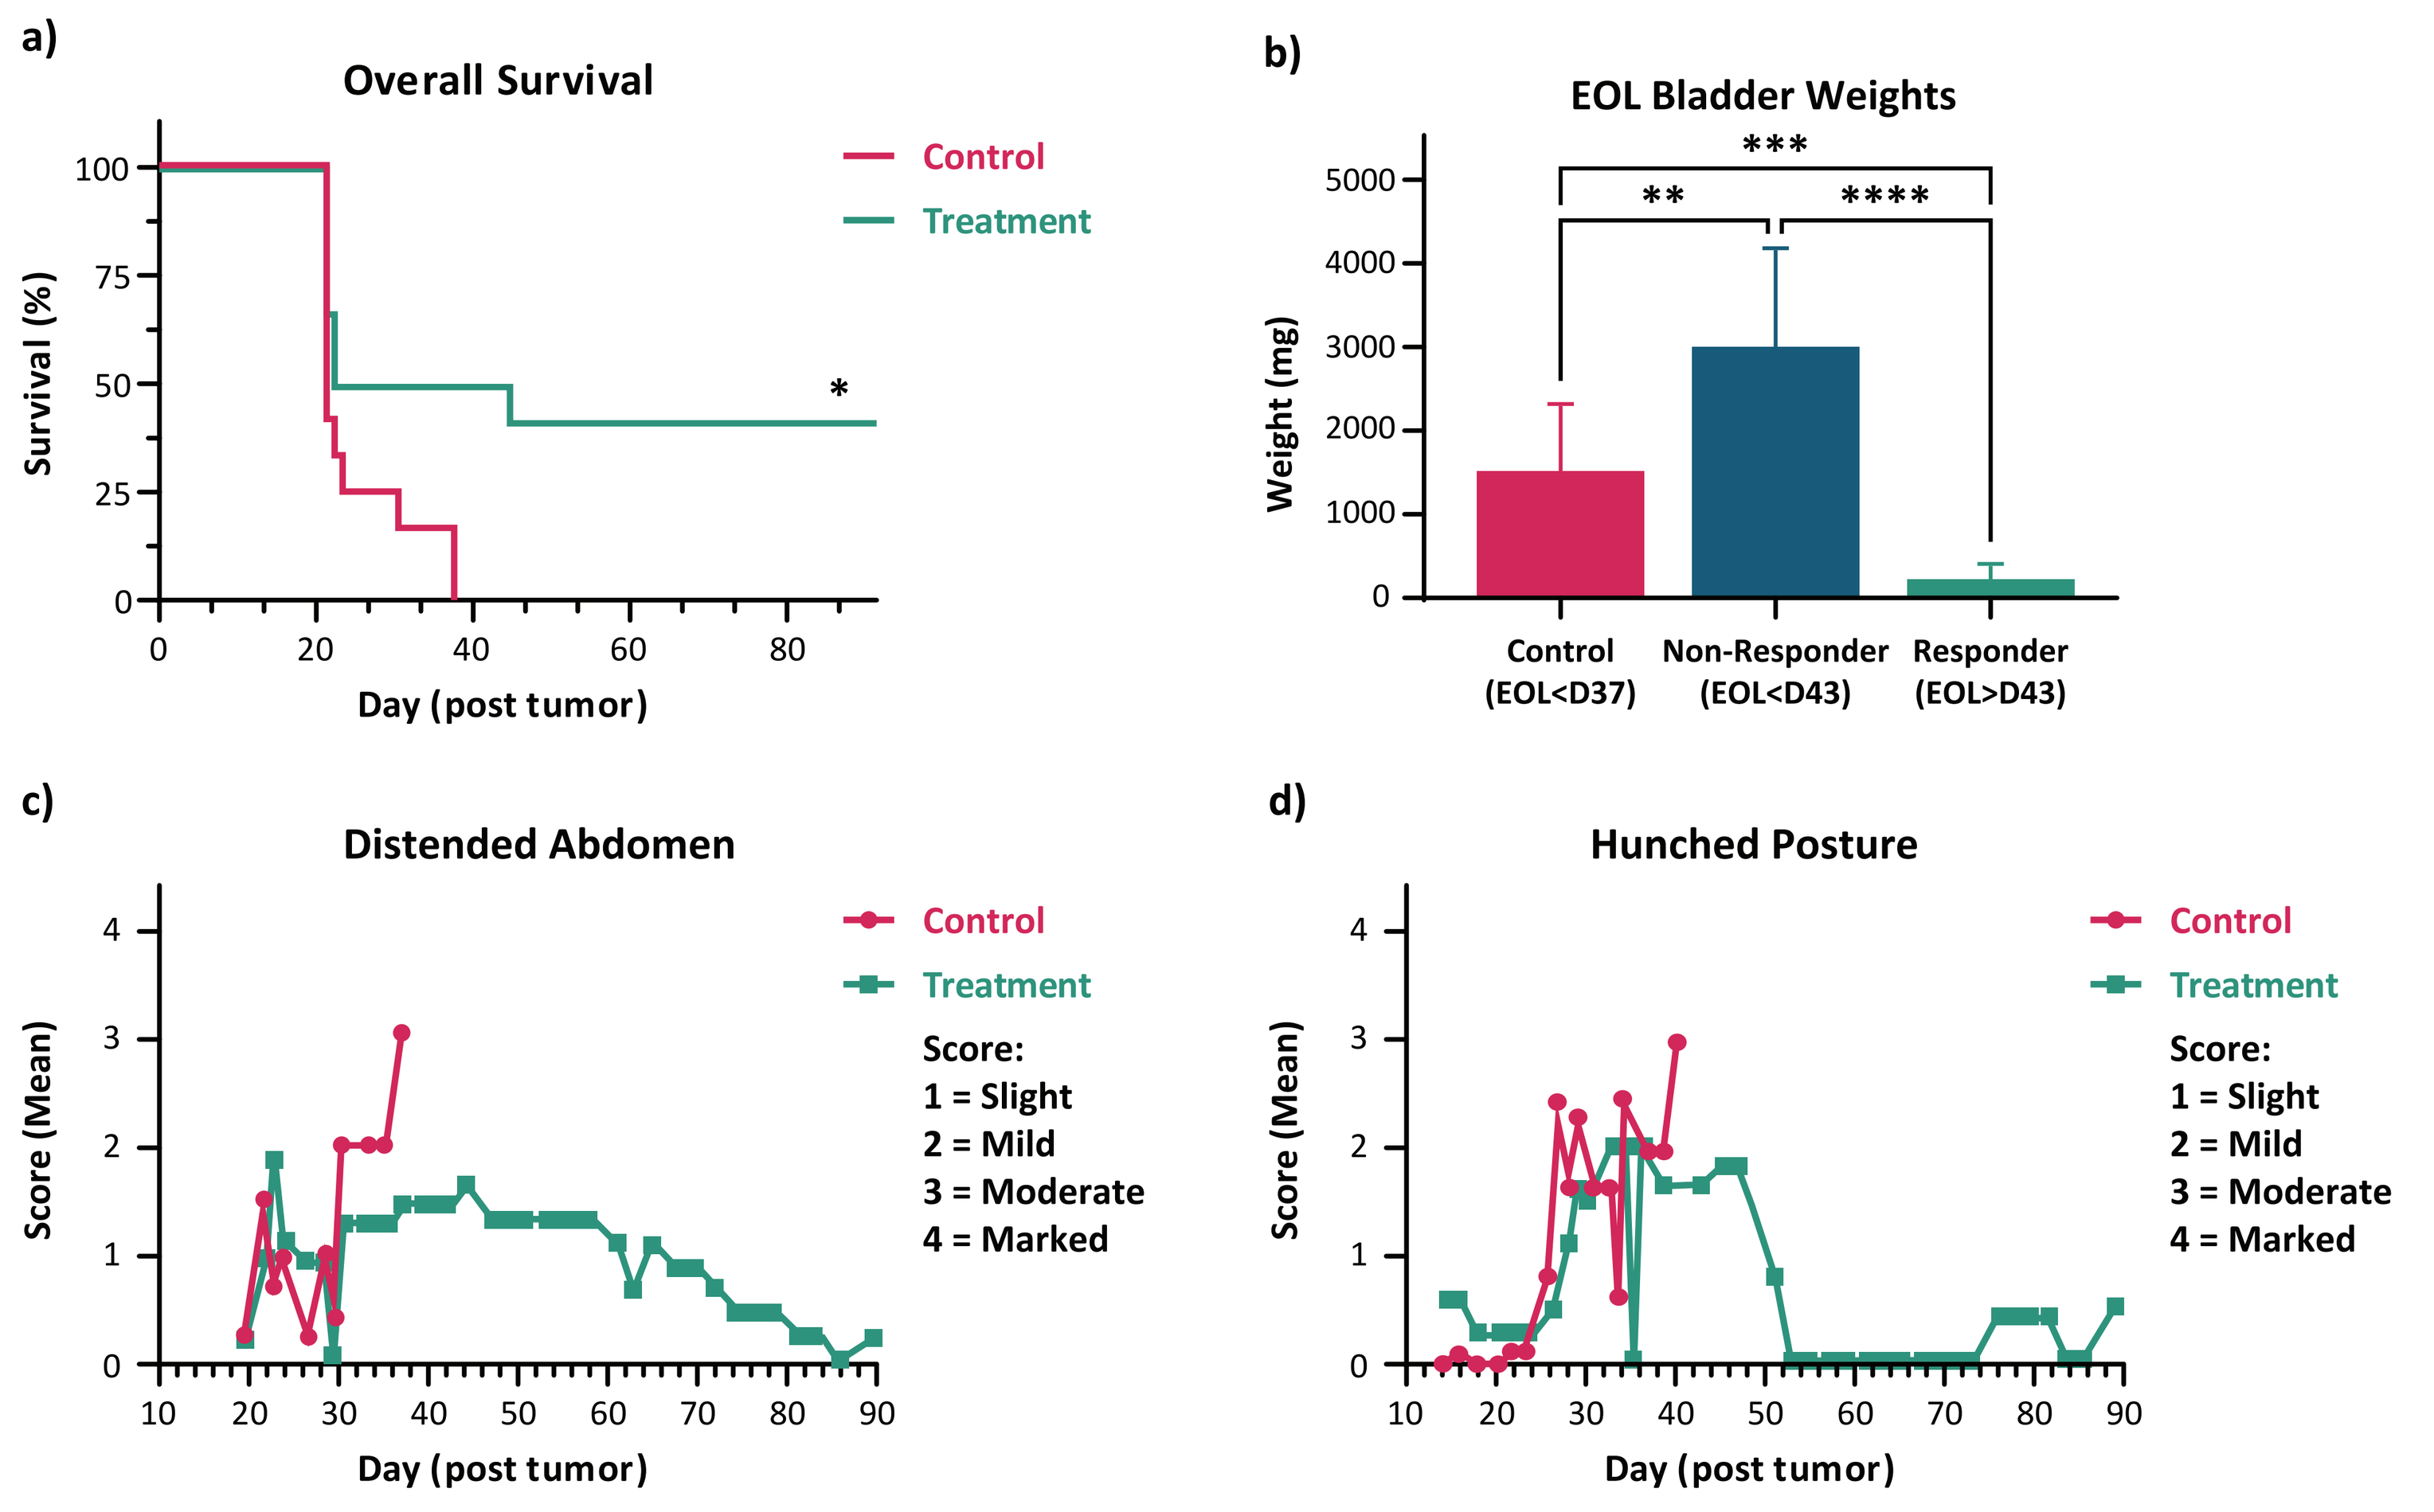
a)
Overall Survival
Survival (%)
Day (post tumor)
Control
Treatment
b)
EOL Bladder Weights
Weight (mg)
Control (EOL<D37)
Non-Responder (EOL<D43)
Responder (EOL>D43)
c)
Distended Abdomen
Score (Mean)
Day (post tumor)
d)
Hunched Posture
Score:
1 = Slight
2 = Mild
3 = Moderate
4 = Marked

Immunotherapy significantly improves **a)** survival and reduces **b)** tumor size in the murine bladder cancer model. Also, it is associated with **c)** reduced distended abdomen and **d)** improved posture (Control n=12; Treatment n=12; Treated Non-Responder n=4; Treated Responder n=6). * P=0.0163, ** P=0.0113, *** P=0.0081, **** P<0.0001.

CONCLUSIONS

- These data support the durable antitumor activity of Imvax's immunotherapeutic platform in multiple cancers (ovarian, liver and bladder) beyond GBM.
- Results suggest that efficacy is associated with a systemic and durable immunological response, resulting in generation of Th1 antitumor cytotoxic T cells.
- Lower PD1 expression in T cells of *m*IHC-001 treated mice suggests reduced T cell exhaustion.
- Future studies will investigate additional biomarkers of antitumor response using further phenotypic evaluation of T cell activation/ exhaustion markers and functional Th1/Th2 responses.

REFERENCES

1. Andrews DW, et al. *Clin Cancer Res*. 2021;27(7):1912-1922.
2. Stupp R, et al. *N Engl J Med*. 2005;352(10):987-996.
3. Zilberberg J, et al. *J Immunother Cancer*. 2021;9:A231.
4. Bae KB, et al. *Anticancer Res*. 2020:40(6):3297-3306.
5. Zhang GL, et al. *J Vis Exp*. 2019:143.
6. Li G, et al. *Small*. 2019:15(25):e1900936.
7. Haugen ØP, et al. *PeerJ*. 2020:8:e8328.

Poster presented at the 37th Annual Meeting of the Society for Immunotherapy of Cancer (SITC 2022). November 8–12, 2022, Boston, MA, USA.

© Copyright 2022 IMVAX®, Inc. All rights reserved.